Rhinovirus (RV) C15, but not A16, induces airway hyper-responsiveness in human small airways and airway smooth muscle (HASM) through modulation of calcium and actin dynamics

Vishal Parikh[1], Riva Patel[1], Corinne Corbi[1], Dylan Marchione[2], Benjamin Garcia[2], Stephen Liggett[3], Reynold A. Panettieri Jr.[1], and Cynthia Koziol-White[1]

[1]Rutgers University, New Brunswick, NJ, [2]University of Pennsylvania, Philadelphia, PA, and [3]University of South Florida, Tampa, FL

Abstract

RATIONALE: Virus-induced respiratory infections, a leading cause of asthma exacerbations, are primarily evoked by rhinovirus. Direct effects of RV on small airways of humans *ex vivo* remains poorly understood. Mechanisms underlying hyperresponsiveness following exposure to RV have not been examined. Therefore, we hypothesized that RV-induced airway hyperrresponsiveness is mediated by modulation of calcium-dependent and –independent pathways.

METHODS: Human precision cut lung slices (hPCLS), each containing a small airway, were exposed to RVA16 or RVC15, and bronchoconstriction carbachol (Cch, 10^{-8} – 10^{-4} M) was measured and compared to no contractile agonist, with maximal bronchoconstriction, area under the curve, and log EC50 of contractile agonist measured from the sigmoidal dose response curves and compared. Intracellular calcium flux was measured by Fluo8 fluorescence in HASM following epithelial cell exposure to RVA16 or C15. Unbiased phosphoproteomic analysis of HASM lysates was performed by LC-MS/MS following phosphopeptide enrichment of the samples.

RESULTS: Airway responsiveness following stimulation with RVC15, but not RVA16, increased AHR in hPCLS (RVC15 compared to control: AUC ANOVA p=0.03; control vs 10^7 Pfu, 136 vs 202, p=0.02; RVA16 vs control, n.s.). RVC15, but not RVA16, elicited greater calcium flux following Cch stimulation in HASM (peak calcium and AUC, p<0.05). RVC15 exposure with Cch stimulation augmented phosphorylation of cytoskeletal-associated proteins including vinculin and HSP27 above Cch stimulation alone.

CONCLUSIONS: These data suggest that differences in contractility following rhinovirus exposure may be a result of changes in calcium dynamics and actin cytoskeletal modulation. Further study is needed to tease apart definitive mechanisms by which RV alters contractility of HASM.

.

Rhinovirus induces airway hyper-responsiveness and inflammation

- Viruses induce wheezing and airway hyper-responsiveness in normal individuals and those with underlying respiratory disorders like asthma.
- In murine models of virus-induced respiratory infection and exacerbations of allergic airway disease, viral exposure elicits airway hyper-responsiveness.
- Direct effects of a viral mimetic in airway smooth muscle elicits mediator production, even in the absence of increased excitation/contraction coupling.
- Rhinovirus serotypes A and C induce more severe symptoms than B serotypes. Rhinovirus C serotypes are known to be associated with the most severe respiratory symptoms observed, as well as the most severe exacerbations of underlying airways diseases, than A or B serotypes.

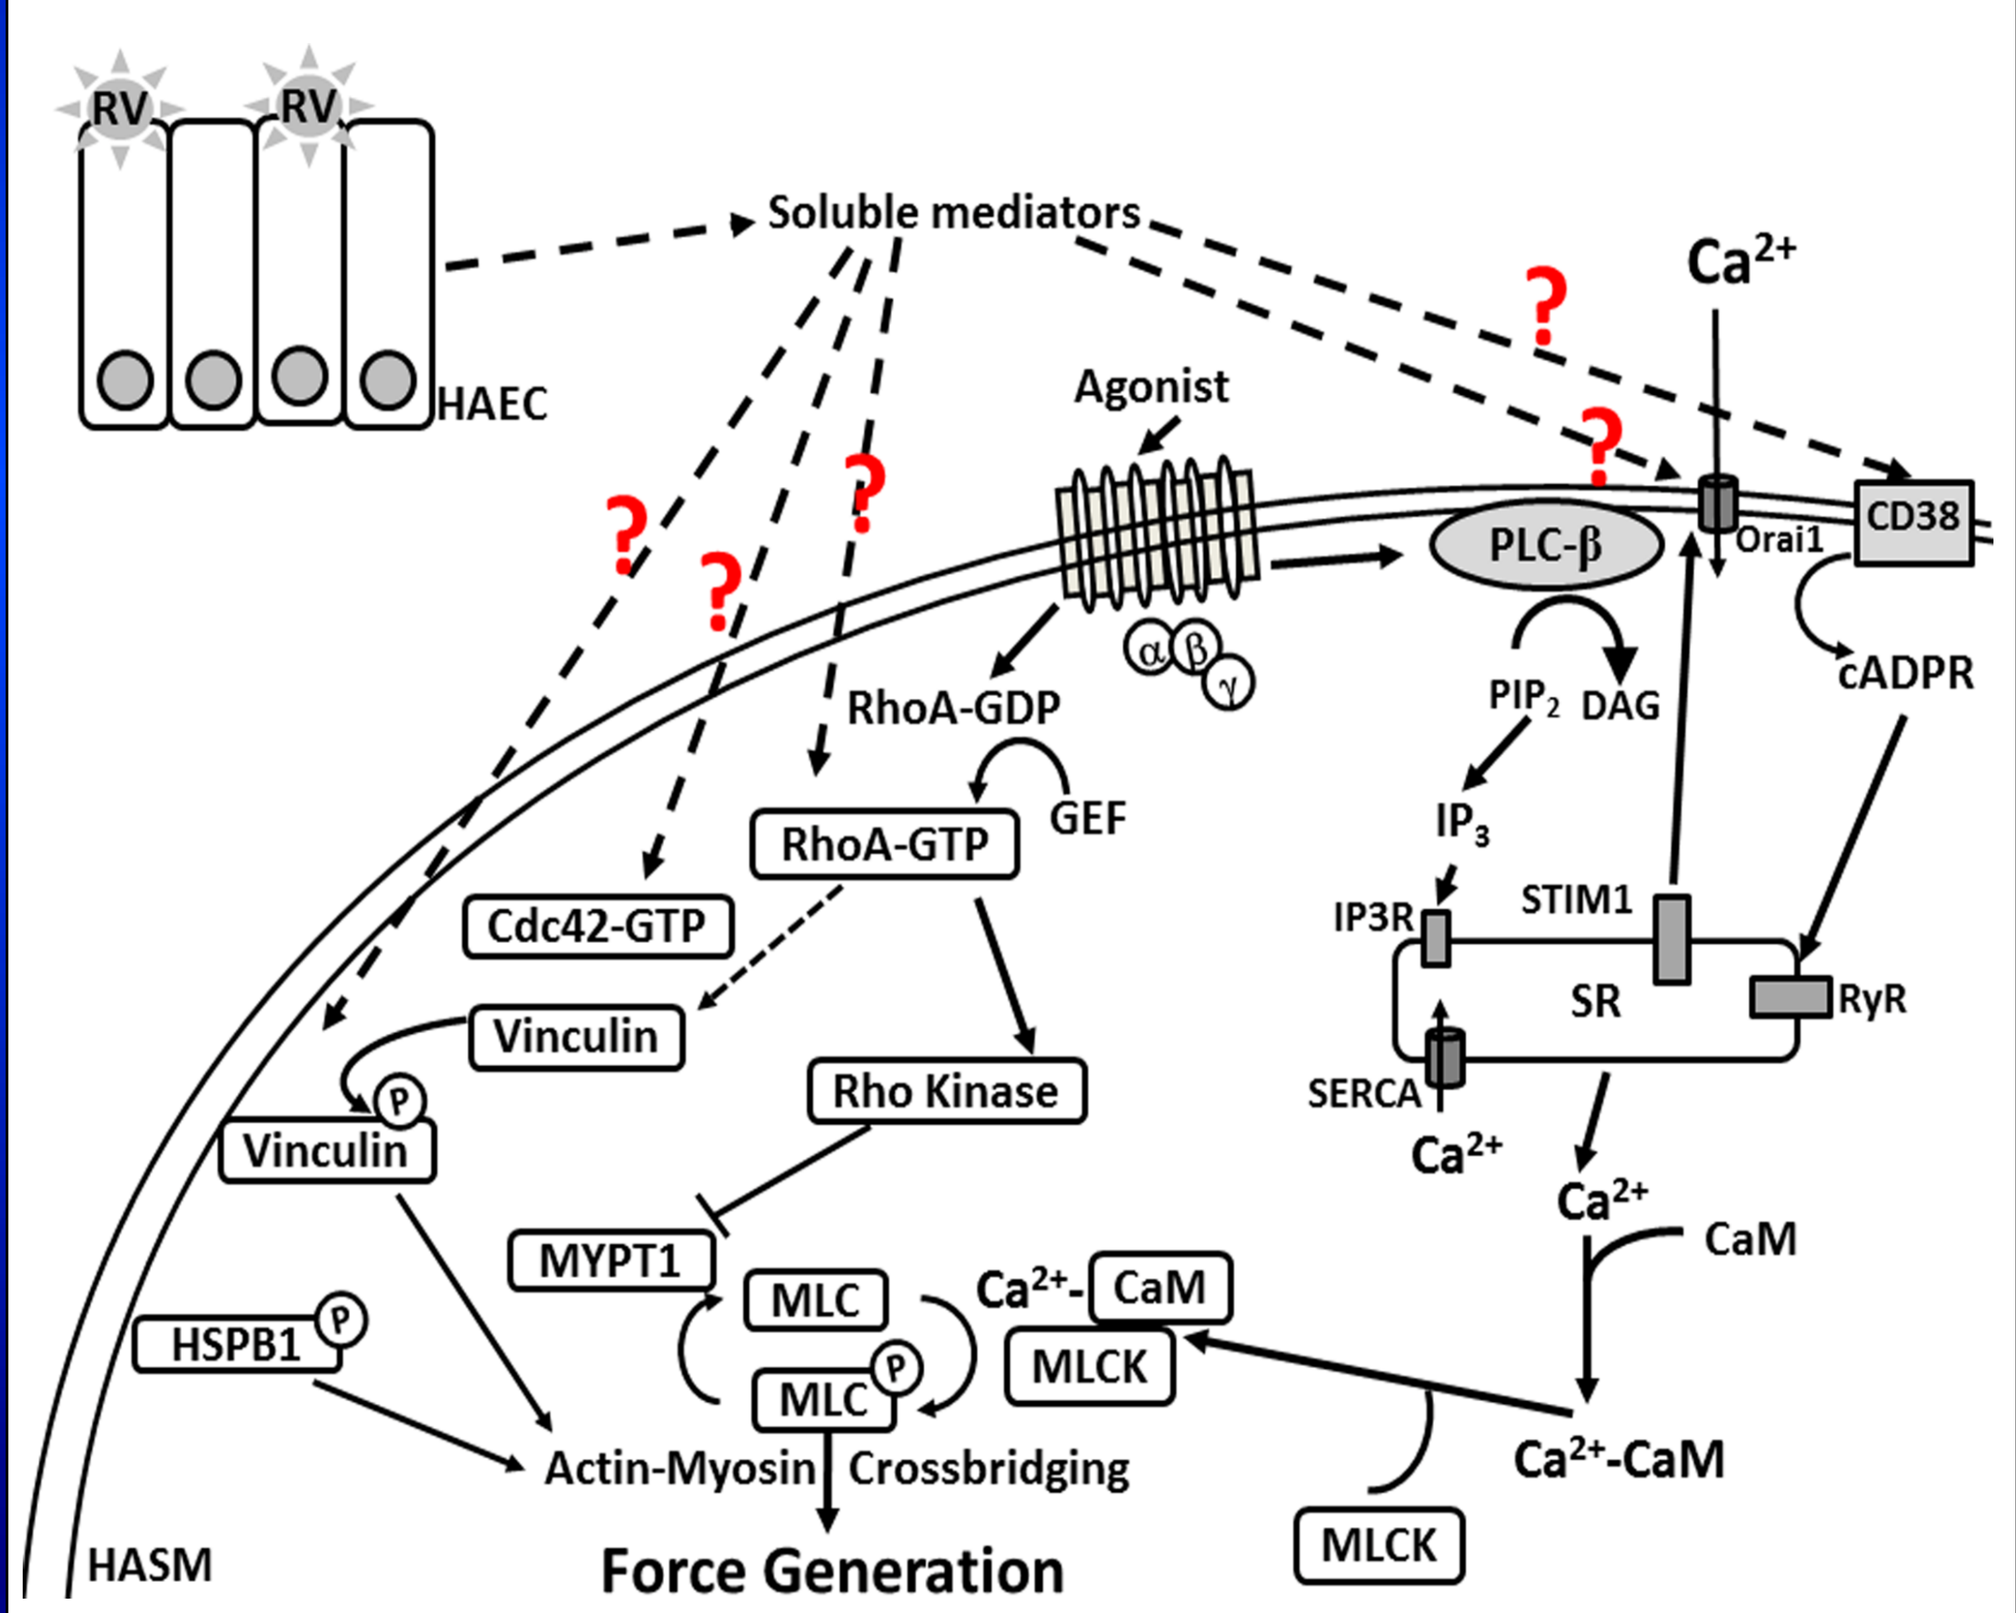
RV
HAEC
Soluble mediators
Agonist
Ca2+
PLC-β
Orai1
CD38
cADPR
RhoA-GDP
GEF
RhoA-GTP
PIP2 DAG
IP3
IP3R
STIM1
SR
RyR
SERCA
Cdc42-GTP
Vinculin
Rho Kinase
CaM
MYPT1
MLC
Ca2+-CaM
MLCK
HSPB1
Actin-Myosin Crossbridging
Force Generation
HASM

Hypothesis

Rhinovirus-induced airway hyperresponsiveness modulates calcium - dependent and - independent pathways in HASM.

RVC15, but not A16, enhances carbachol-induced human small airway constriction

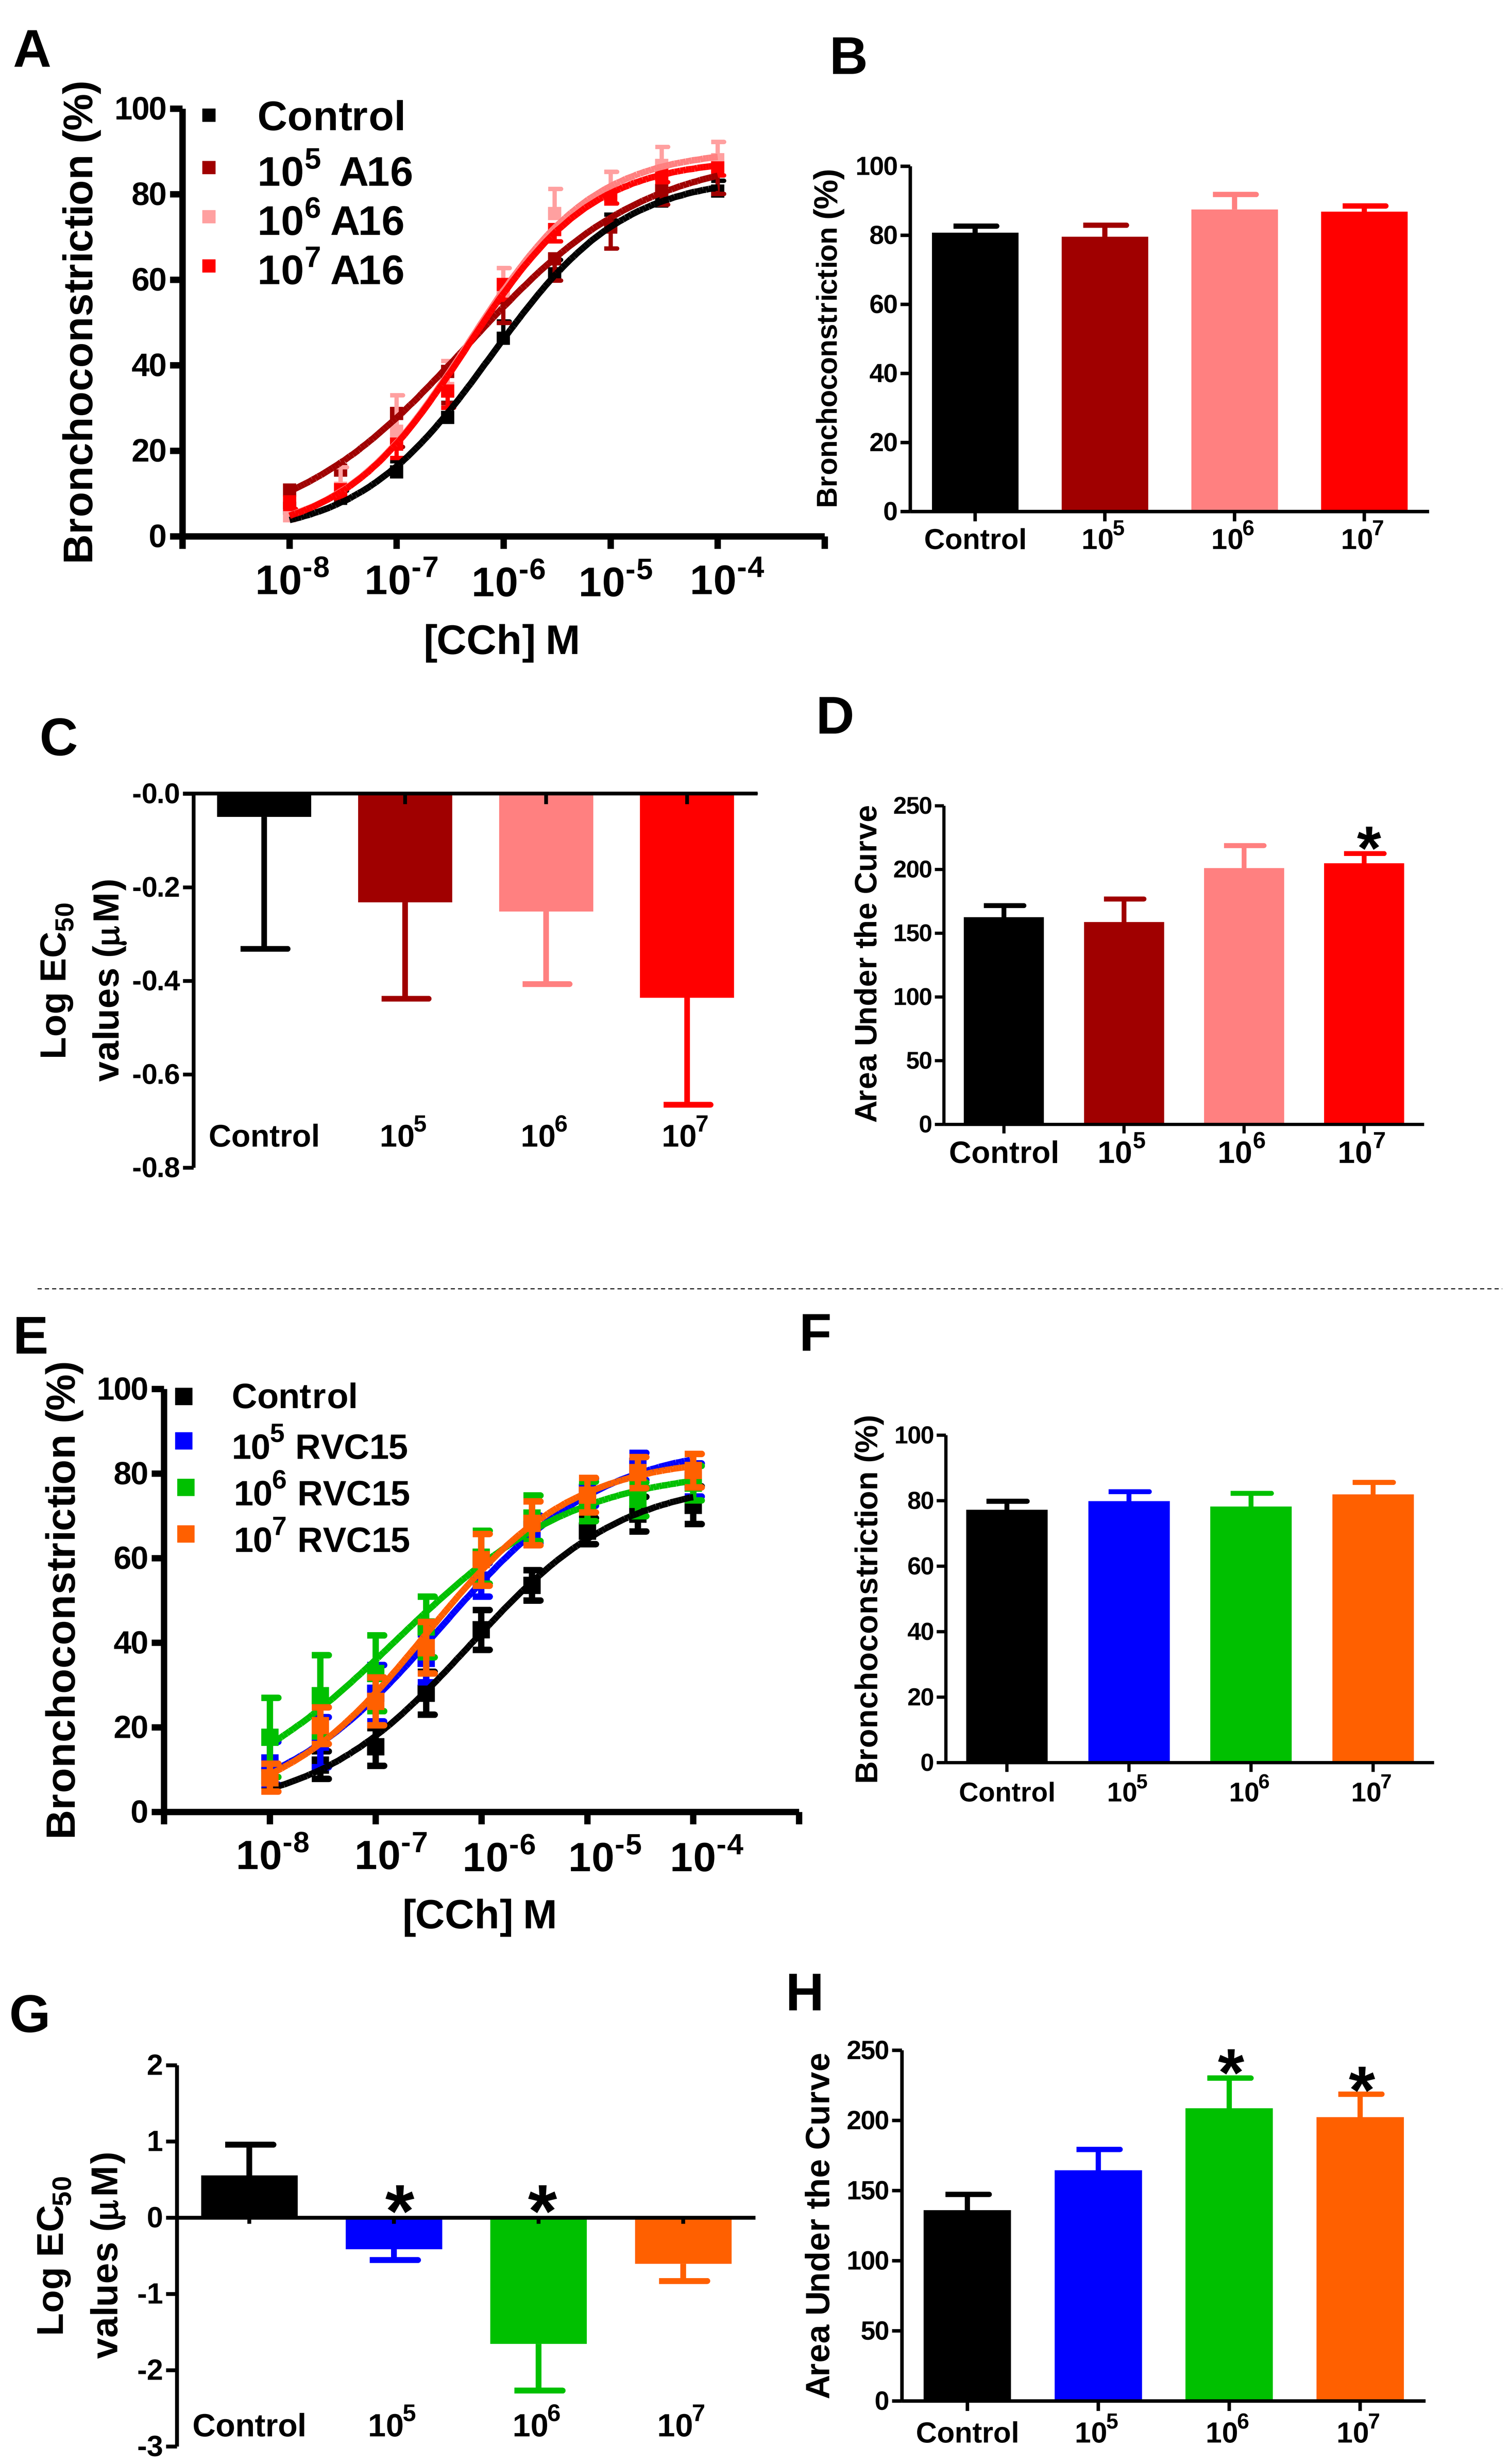
A
Bronchoconstriction (%)
Control
10^5 A16
10^6 A16
10^7 A16
[CCh] M
B
C
Log EC50 values (µM)
D
Area Under the Curve
E
Control
10^5 RVC15
10^6 RVC15
10^7 RVC15
F
G
H

Figure 1 – RVC15, and to a lesser extent RVA16, increases responsiveness of small airways to carbachol in hPCLS. Human precision cut lung slices (hPCLS) were exposed to a dose response of (A-D) RVA16 or (E-H) RVC15 (48 hr). Agonist-induced airway narrowing was measured to a dose response of carbachol. RVA16:n≥18 separate donors, AUC ANOVA p=0.0045. RVC15:n≥10 separate donors, *p≤0.05 vs control buffer.

RVC15, but not A16, augments carbachol-induced calcium levels in ALI-differentiated epithelial/HASM cell co-cultures

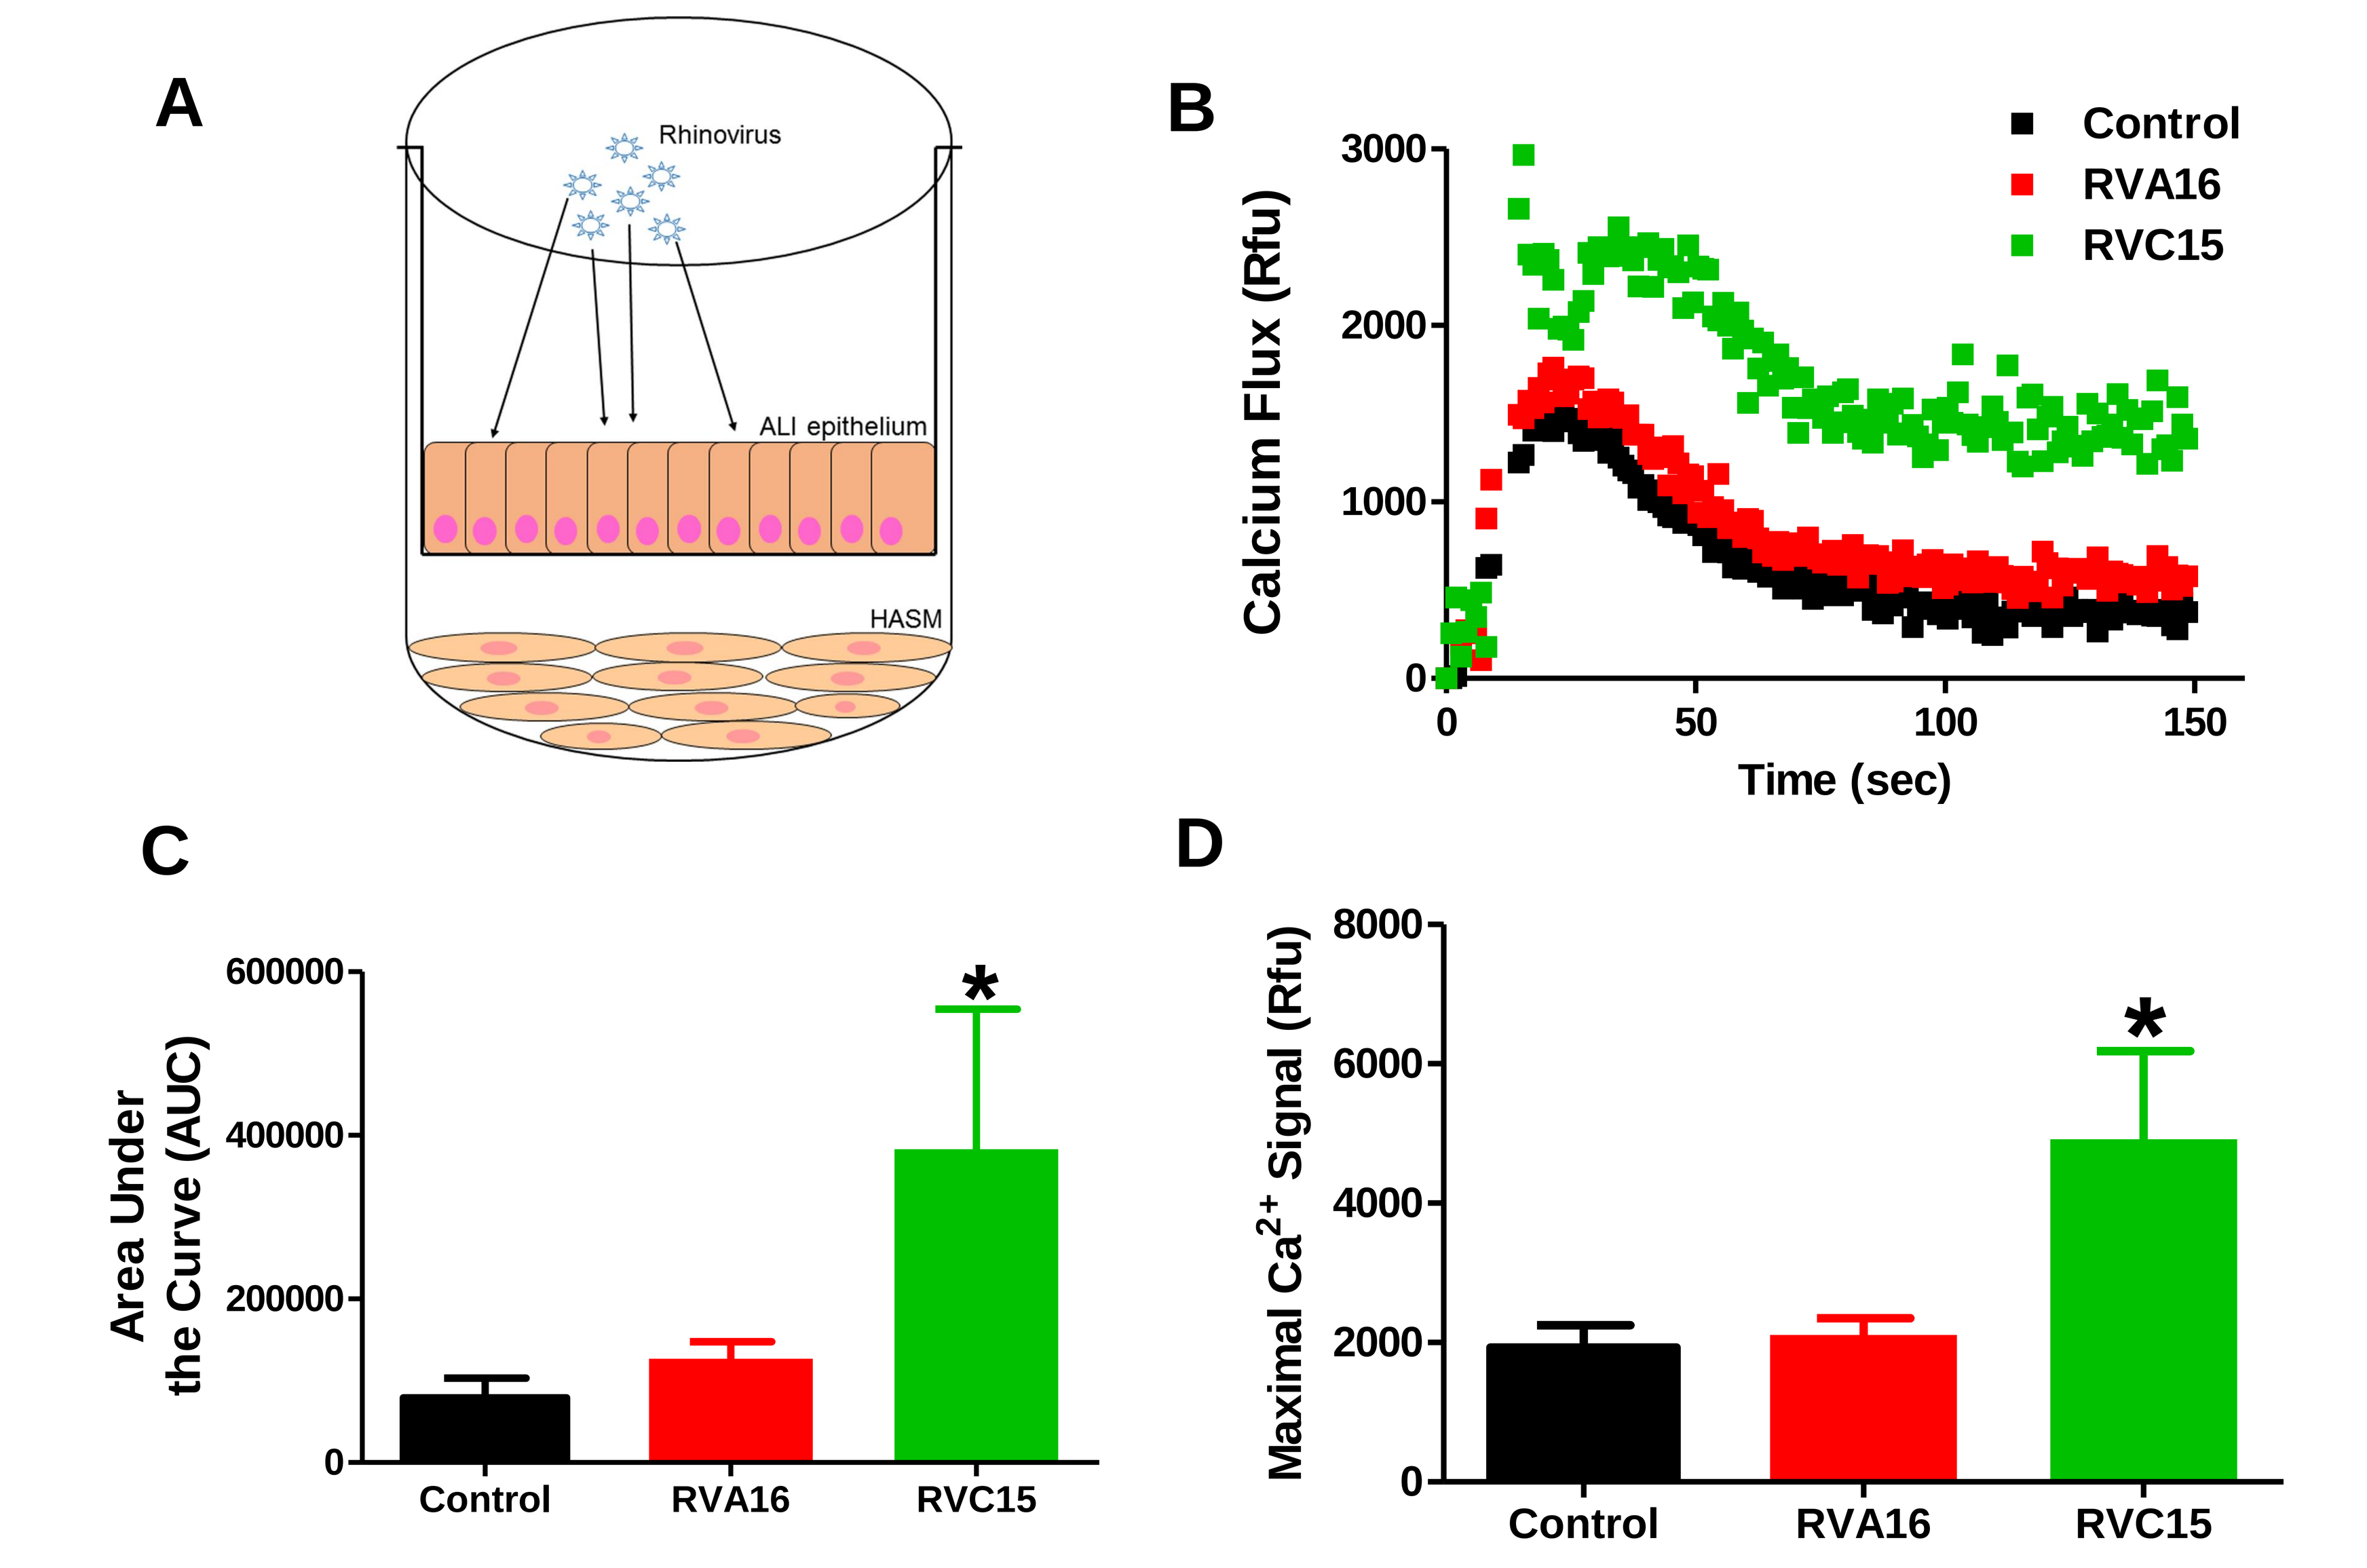
A
Rhinovirus
ALI epithelium
HASM
B
Calcium Flux (Rfu)
Time (sec)
Control
RVA16
RVC15
C
Area Under the Curve (AUC)
D
Maximal Ca2+ Signal (Rfu)

Figure 2 – RVC15-exposed ALI-differentiated epithelial cells induce increased calcium transients in co-cultured HASM, with RVA16 having little effect on calcium transients. Epithelial cells were exposed to RV16 (10^7 Pfu, 48 hr) cultured above HASM (A). Bulk calcium transients to carbachol were measured using Fluo-8 over 120 sec (B). Exposure to RV alone did not induce calcium flux. Area under the curve (C) and maximal calcium (D - relative fluorescence units) were calculated and plotted as mean ± standard error. * p<0.05, n=3 distinct donor cell co-culture experiments.

RVC15 induces phosphorylation of proteins associated with actin cytoskeletal rearrangement

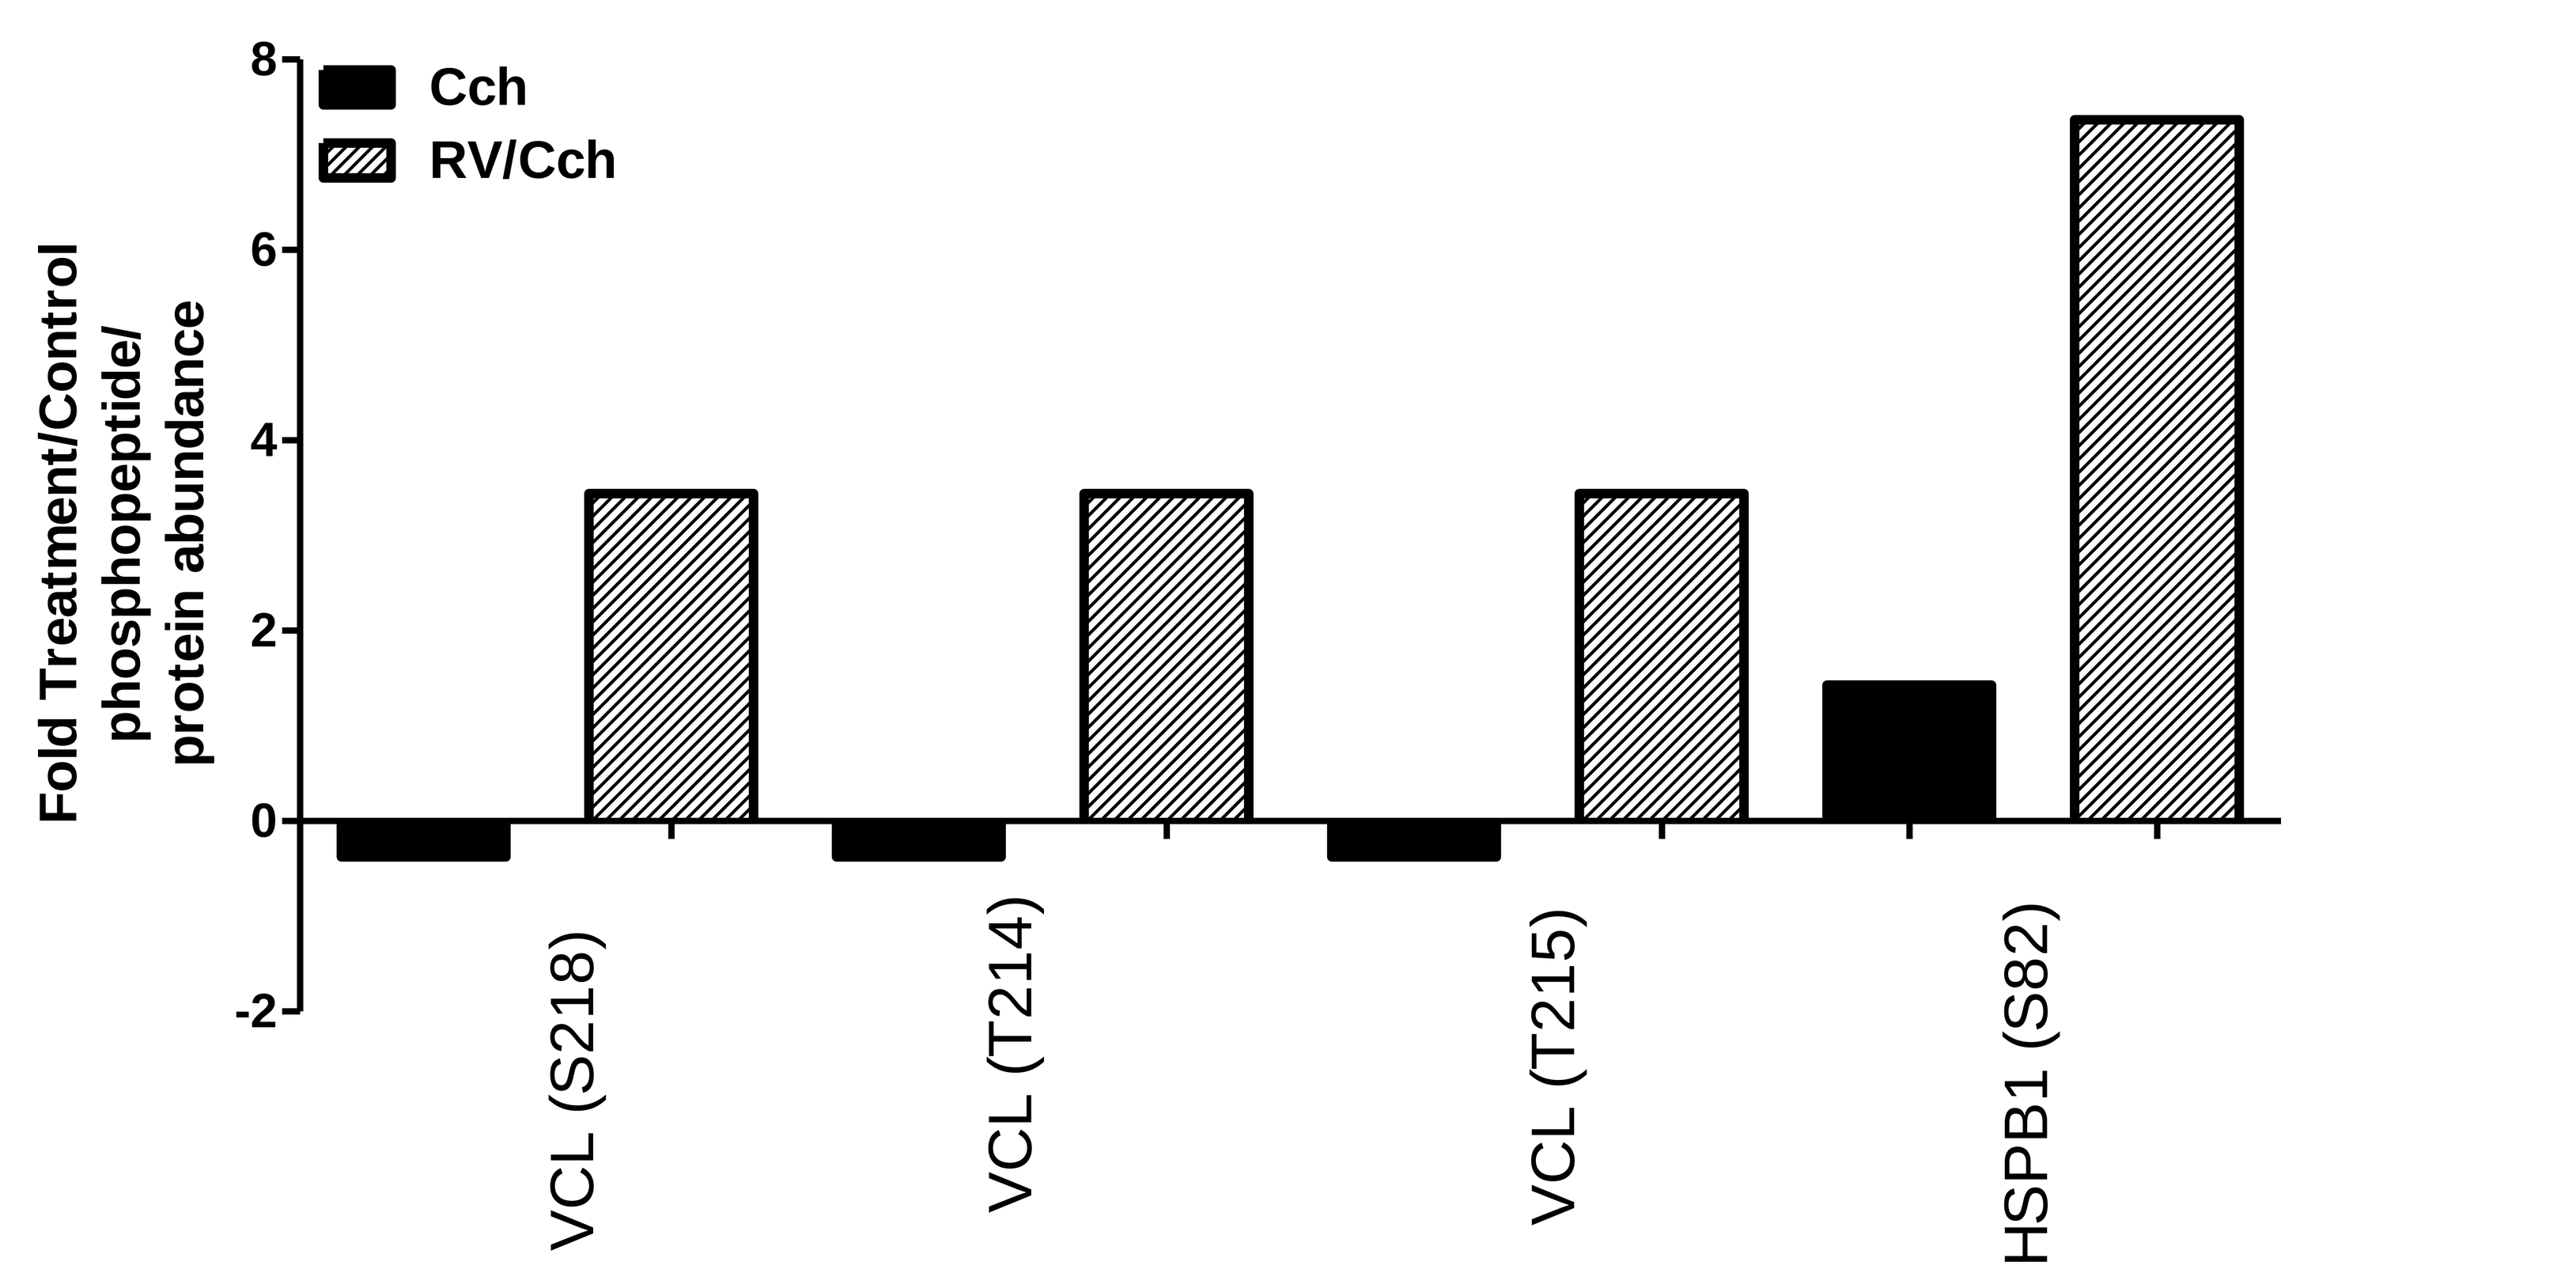
Cch
RV/Cch
Fold Treatment/Control phosphopeptide/ protein abundance
VCL (S218)
VCL (T214)
VCL (T215)
HSPB1 (S82)

Figure 3 – RV15 exposure of ALI-differentiated epithelial cells augments phosphorylation of actin cytoskeleton-associated proteins following carbachol exposure of co-culture HASM. Non-diseased ALI HAECs were stimulated with RVC15 (10^7 Pfu, 48 hr) with HASM from the same donor co-cultured in a plate below. The HASM were stimulated with +/- carbachol (Cch, 10 µM – 10 min), lysed, and processed for mass spec. Phosphopeptides are denoted as fold compared to control buffer/basal for either RV exposure alone or RV exposure with subsequent Cch stimulation.

Summary

- Exposure of RVC15, but not A16, augments carbachol-induced bronchoconstriction by affecting the sensitivity of the airways as well as affecting the integrated response following contractile agonist stimulation.
- Co-cultures of ALI-differentiated epithelial cells (HAEC) showed enhanced responsiveness of HASM cells to carbachol-induced calcium flux following exposure to RVC15, but not RVA16.
- RVC15 exposure of HAEC/HASM co-cultures augments carbachol-induced phosphorylation of actin cytoskeleton-associated proteins (vinculin and HSPB1).

Significance

Defining mechanisms by which rhinovirus modulates contractility of smooth muscle will provide novel therapeutic targets to abrogate exacerbations of asthma.

References

1. Cooper PR *et al.* (2003) TLR3 activation stimulates cytokine secretion without altering agonist-induced human small airway contraction or relaxation. *Am J Physiol Lung Cell Mol Physiol. 297:L530*.
2. Clarke D *et al.* (2009) Transcriptional regulation of cytokine function in airway smooth muscle cells. *Pulm Pharmacol Ther.* 22:436.
3. Oliver BB *et al.* (2006) Increased proinflammatory responses from asthmatic airway smooth muscle cells in response to rhinovirus infection. *Respir Res*. 7:71.
4. Bizzintino J *et al.* (2011) Association between human rhinovirus C and severity of acute asthma in children. *Eur Respir J*. 37:1037.
5. Jackson DJ *et al.* (2008) Wheezing rhinovirus illnesses in early life predict asthma development in high-risk children. *Am J Respir Crit Care Med*. 178:667.
6. Palmenberg AC *et al.* (2009) Sequencing and analyses of all known human rhinovirus genomes reveals structure and evolution. *Science*. 324:55.
7. Kennedy JL, Koziol-White CJ, *et al.* (2018) Effects of rhinovirus 39 infection on airway hyperresponsiveness to carbachol in human airways precision cut lung slices. *JACI*. Epub ahead of print.

These studies were supported by NIH P01-HL114471-03