TGF-β1 Mediates HASM Cell Hyporesponsiveness to β2-Agonist-Induced Relaxation

Christie A. Ojiaku[1,2], Jazmean K. Williams[3], Sam Paek[4], Shreya Narayan[4], Rennolds S Ostrom[5], Steven S. An[4], and Reynold A. Panettieri Jr.[2]

[1]Department of Systems Pharmacology and Translational Therapeutics, University of Pennsylvania, Philadelphia, PA
[2]Rutgers Institute for Translational Medicine and Science, Rutgers, The State University of New Jersey, New Brunswick, NJ
[3]Drexel University, Philadelphia, PA
[4]Johns Hopkins Bloomberg School of Public Health, Johns Hopkins University, Baltimore, MD
[5]Department of Biomedical and Pharmaceutical Sciences, Chapman University School of Pharmacy, Irvine, California

ABSTRACT

RATIONALE: Several cytokines modulate both HASM cell agonist-induced hyperresponsiveness and β2-agonist hyporesponsiveness in asthma. Whether transforming growth factor beta 1 (TGF-β1), which induces HASM cell shortening and hyperresponsiveness, modulates HASM cell relaxation remains unclear. We hypothesize that TGF-β1-induced HASM cell shortening is resistant to β2-agonist-mediated relaxation.

METHODS: Human precision-cut lung slices (hPCLS) were treated with TGF-β1 (100 ng/mL) overnight and constricted to >50% with the contractile agonist carbachol. Subsequently, bronchodilation to formoterol was detected in slices by analyzing changes in airway lumen area using a live-feed microscope and an Image Pro-Plus software macro. Serum-starved HASM cells were treated with TGF-β1 (10 ng/mL) overnight prior to stimulation with isoproterenol (ISO) (20 μM, 5 min) in the presence or absence of carbachol (Cch) (20 μM, 10 min). HASM cell protein was isolated and expression of phosphorylated myosin light chain (MLC), AKT, and ERK were determined by immunoblot. Intracellular cAMP levels were determined by chemiluminescent immunoassay in TGF-β1-treated cells following ISO (1 μM, 10 min) and/or Cch (20 μM, 12 min) stimulation.

RESULTS: Overnight TGF-β1 treatment induced a slight reduction in hPCLS airway lumen bronchodilation over that of control. ISO stimulation of HASM cells significantly decreased HASM cell MLC phosphorylation induced by Cch (48.85% ± 22.26; P=0.02), but not that induced by TGF-β1 (26.79% ± 17.1; P=0.44). Additionally, Cch stimulation following overnight TGF-β1 treatment induced a hyperresponsive phenotype in HASM cells that conferred resistance to ISO-induced MLC dephosphorylation. In TGF-β1 treated cells, ISO-induced intracellular cAMP levels were significantly decreased versus that of control. Furthermore, inhibition of phosphodiesterase activity with IBMX (500 μM) reduced the inhibitory effect of TGF-β1 on ISO-induced cAMP generation.

CONCLUSION: Our data suggest that TGF-β1 induces HASM cell β2-agonist hyporesponsiveness by modulating intracellular cAMP levels. The reversal of TGF-β1's inhibitory effect on ISO-induced cAMP with IBMX suggests a role for phosphodiesterases in TGF-β1-induced modulation of HASM cell relaxation.

TGF-β1 in HASM Cell Contractile Responses in Asthma

- The mechanisms governing HASM cell contractile responses in asthma are incompletely understood.
- TGF-β1 - a pro-fibrotic cytokine implicated in asthma[1] – induces HASM cell shortening and airway hyperresponsiveness[2]
- TGF-β1 modulation of HASM cell relaxation pathways may contribute to β2AR hyporesponsiveness and increased HASM cell shortening.

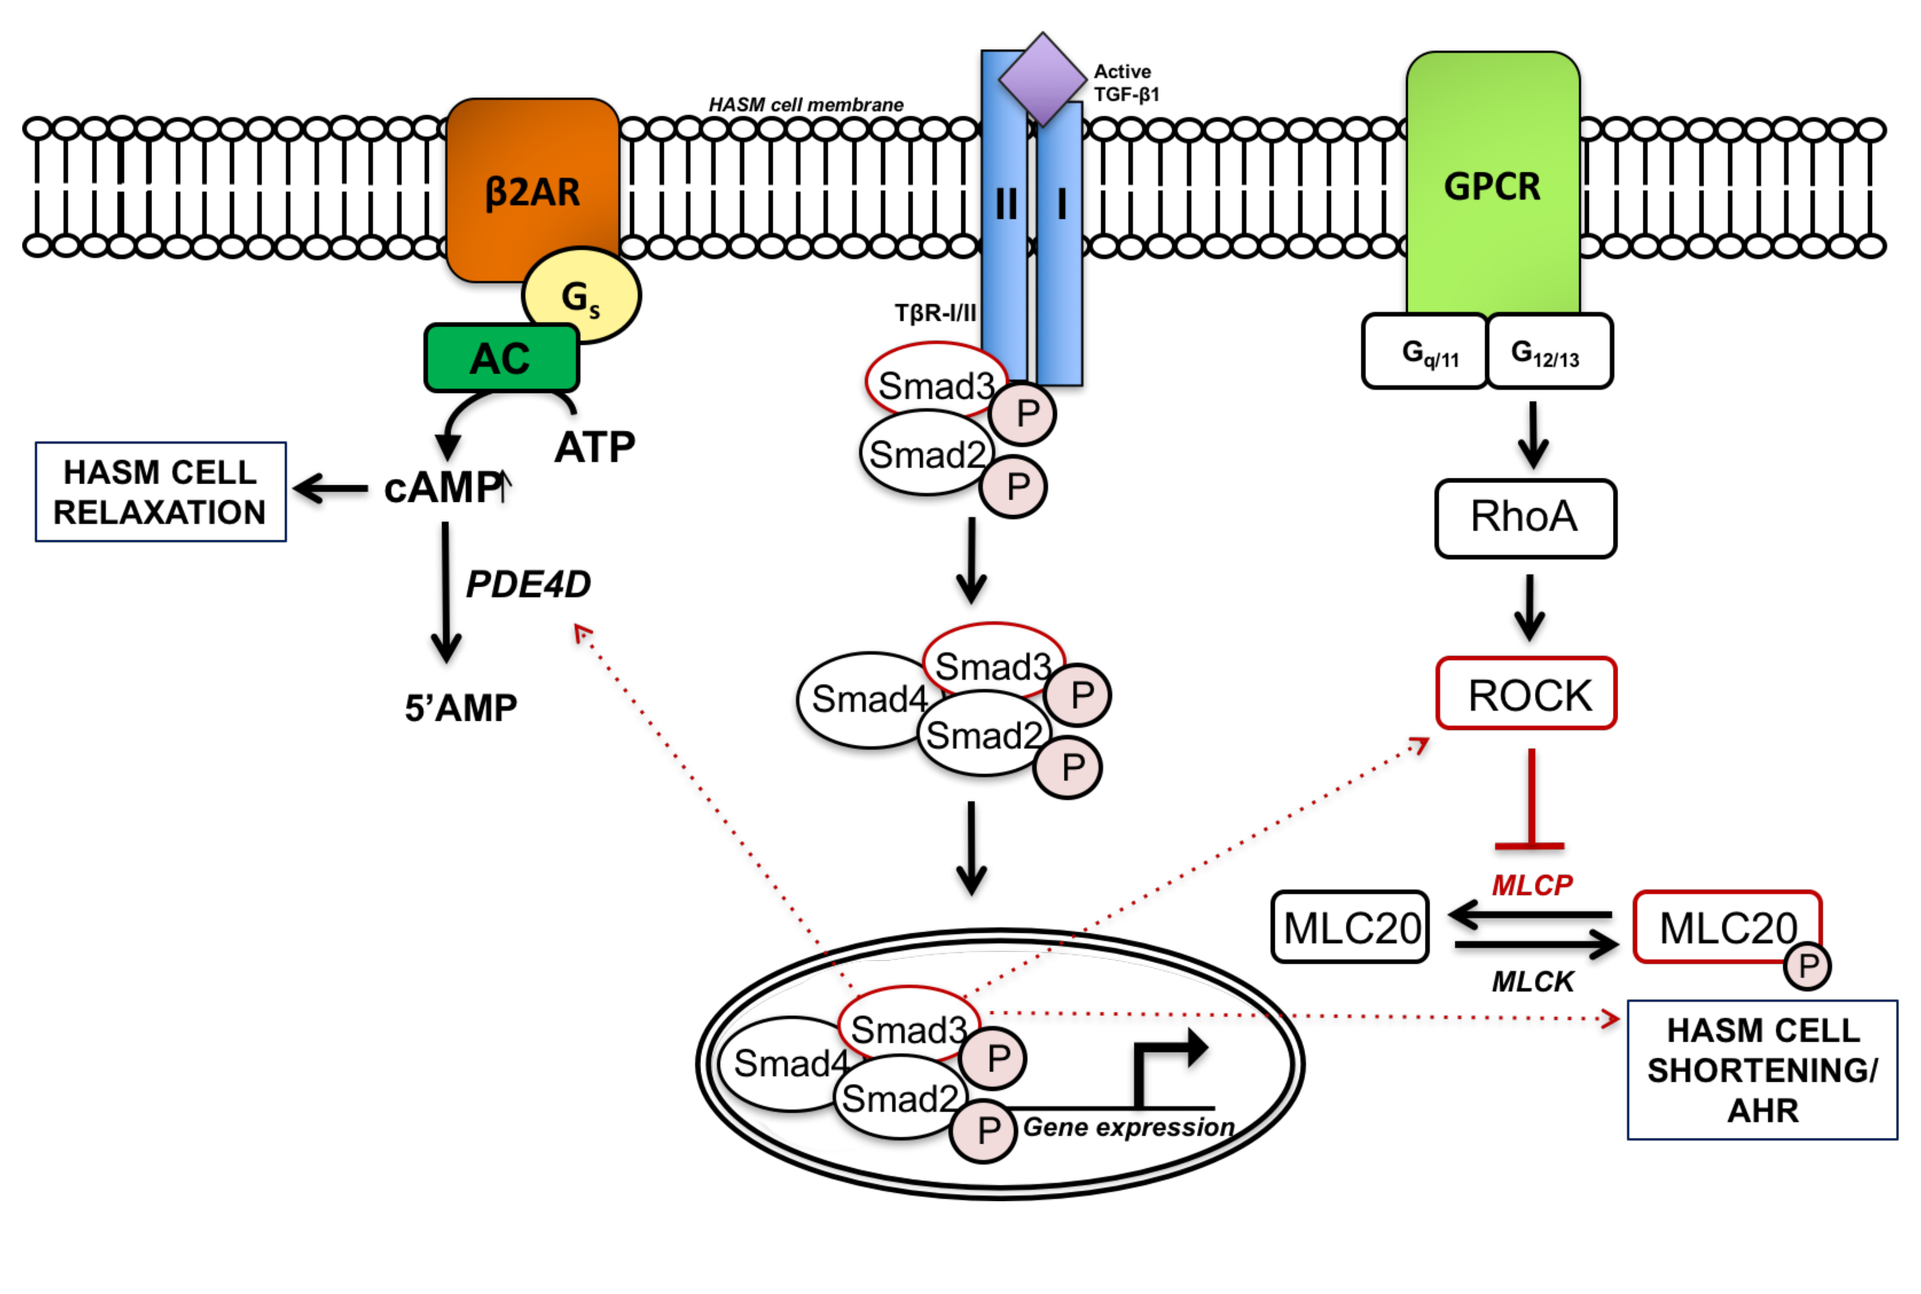

HASM cell membrane
β2AR
AC
ATP
HASM CELL RELAXATION
cAMP
PDE4D
5'AMP
Active TGF-β1
TβR-I/II
Smad3
Smad2
Smad4
P
GPCR
RhoA
ROCK
MLCP
MLCK
MLC20
HASM CELL SHORTENING/ AHR
Gene expression

HYPOTHESIS

TGF-β1 signaling modulates HASM cell relaxation pathways to induce β2-agonist hyporesponsiveness in asthma.

TGF-β1 Inhibits HASM Cell Responsiveness to β2-Adrenergic Receptor (β2AR) Stimuli

β2-Agonist-Induced Inhibition of MLC Phosphorylation

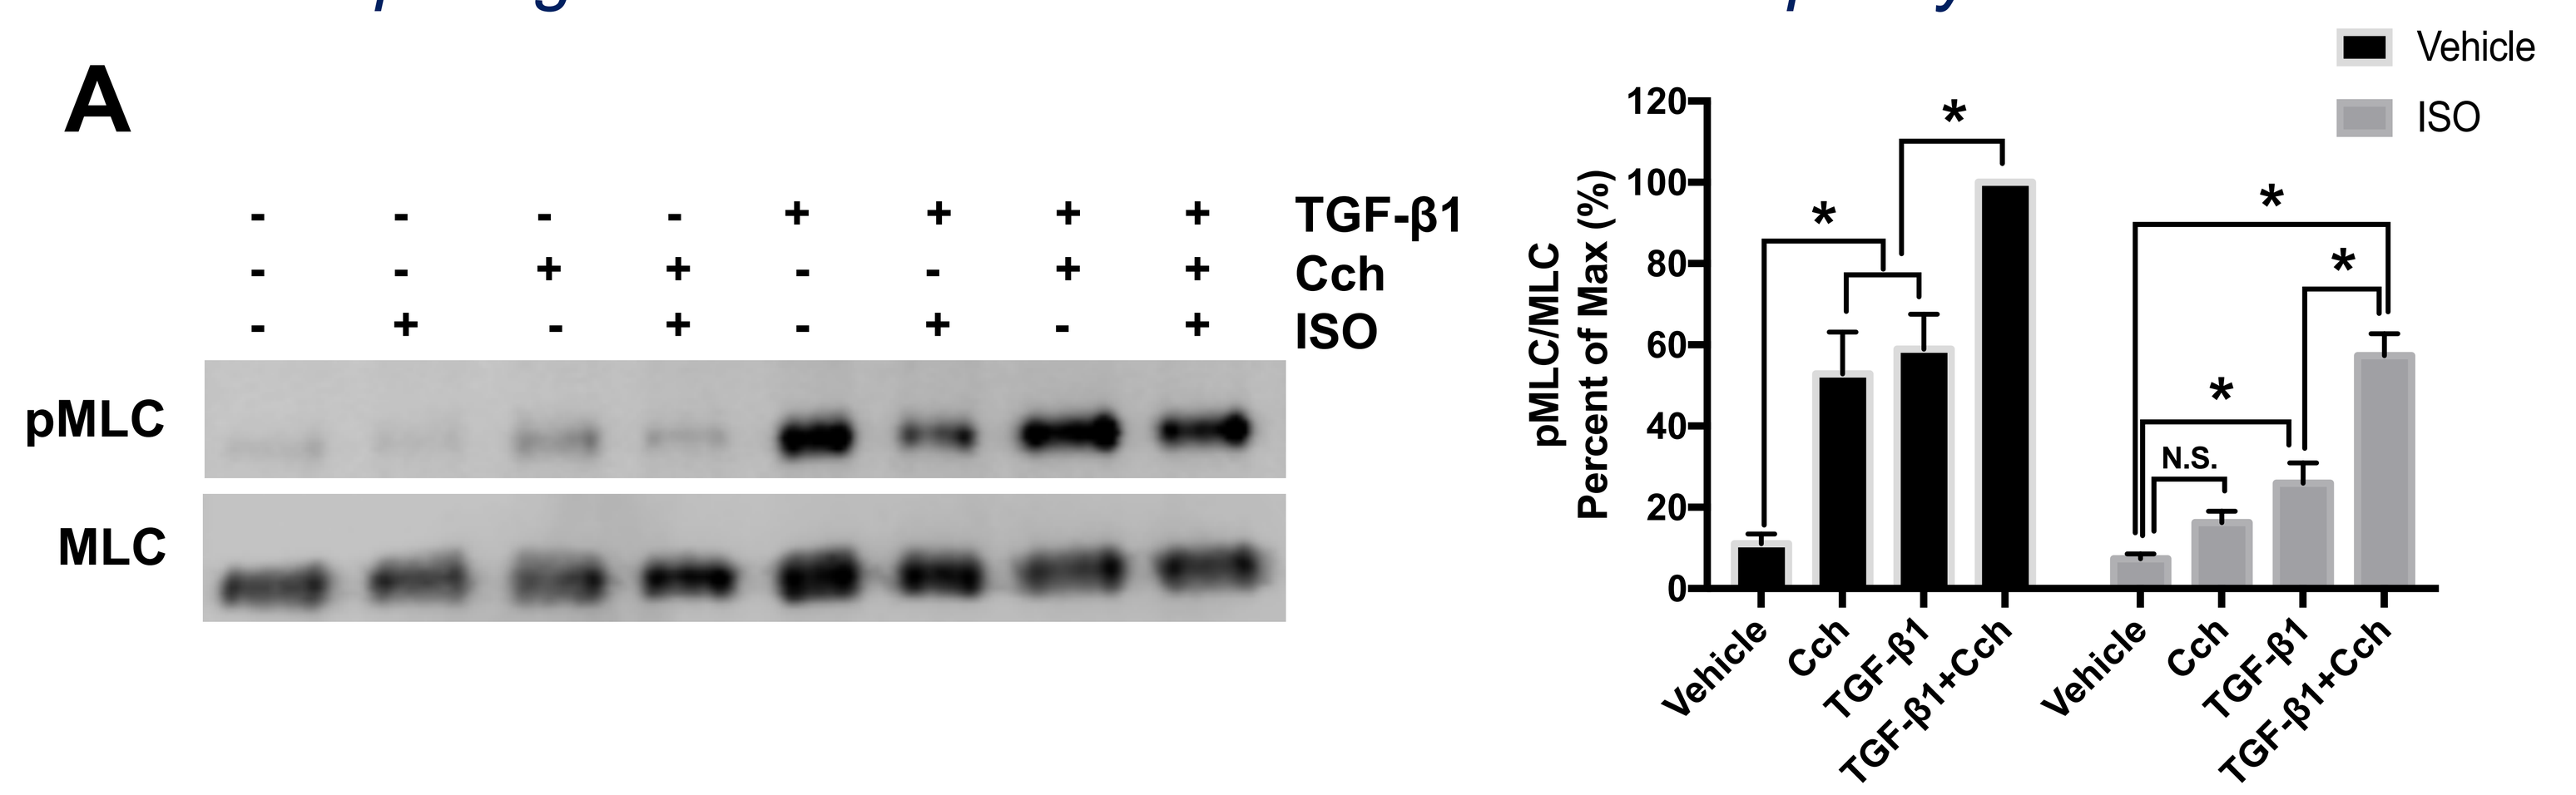

A
TGF-β1
Cch
ISO
pMLC
MLC
pMLC/MLC Percent of Max (%)
Vehicle
ISO
N.S.
Vehicle
Cch
TGF-β1
TGF-β1+Cch

β2-Agonist-Induced cAMP Production

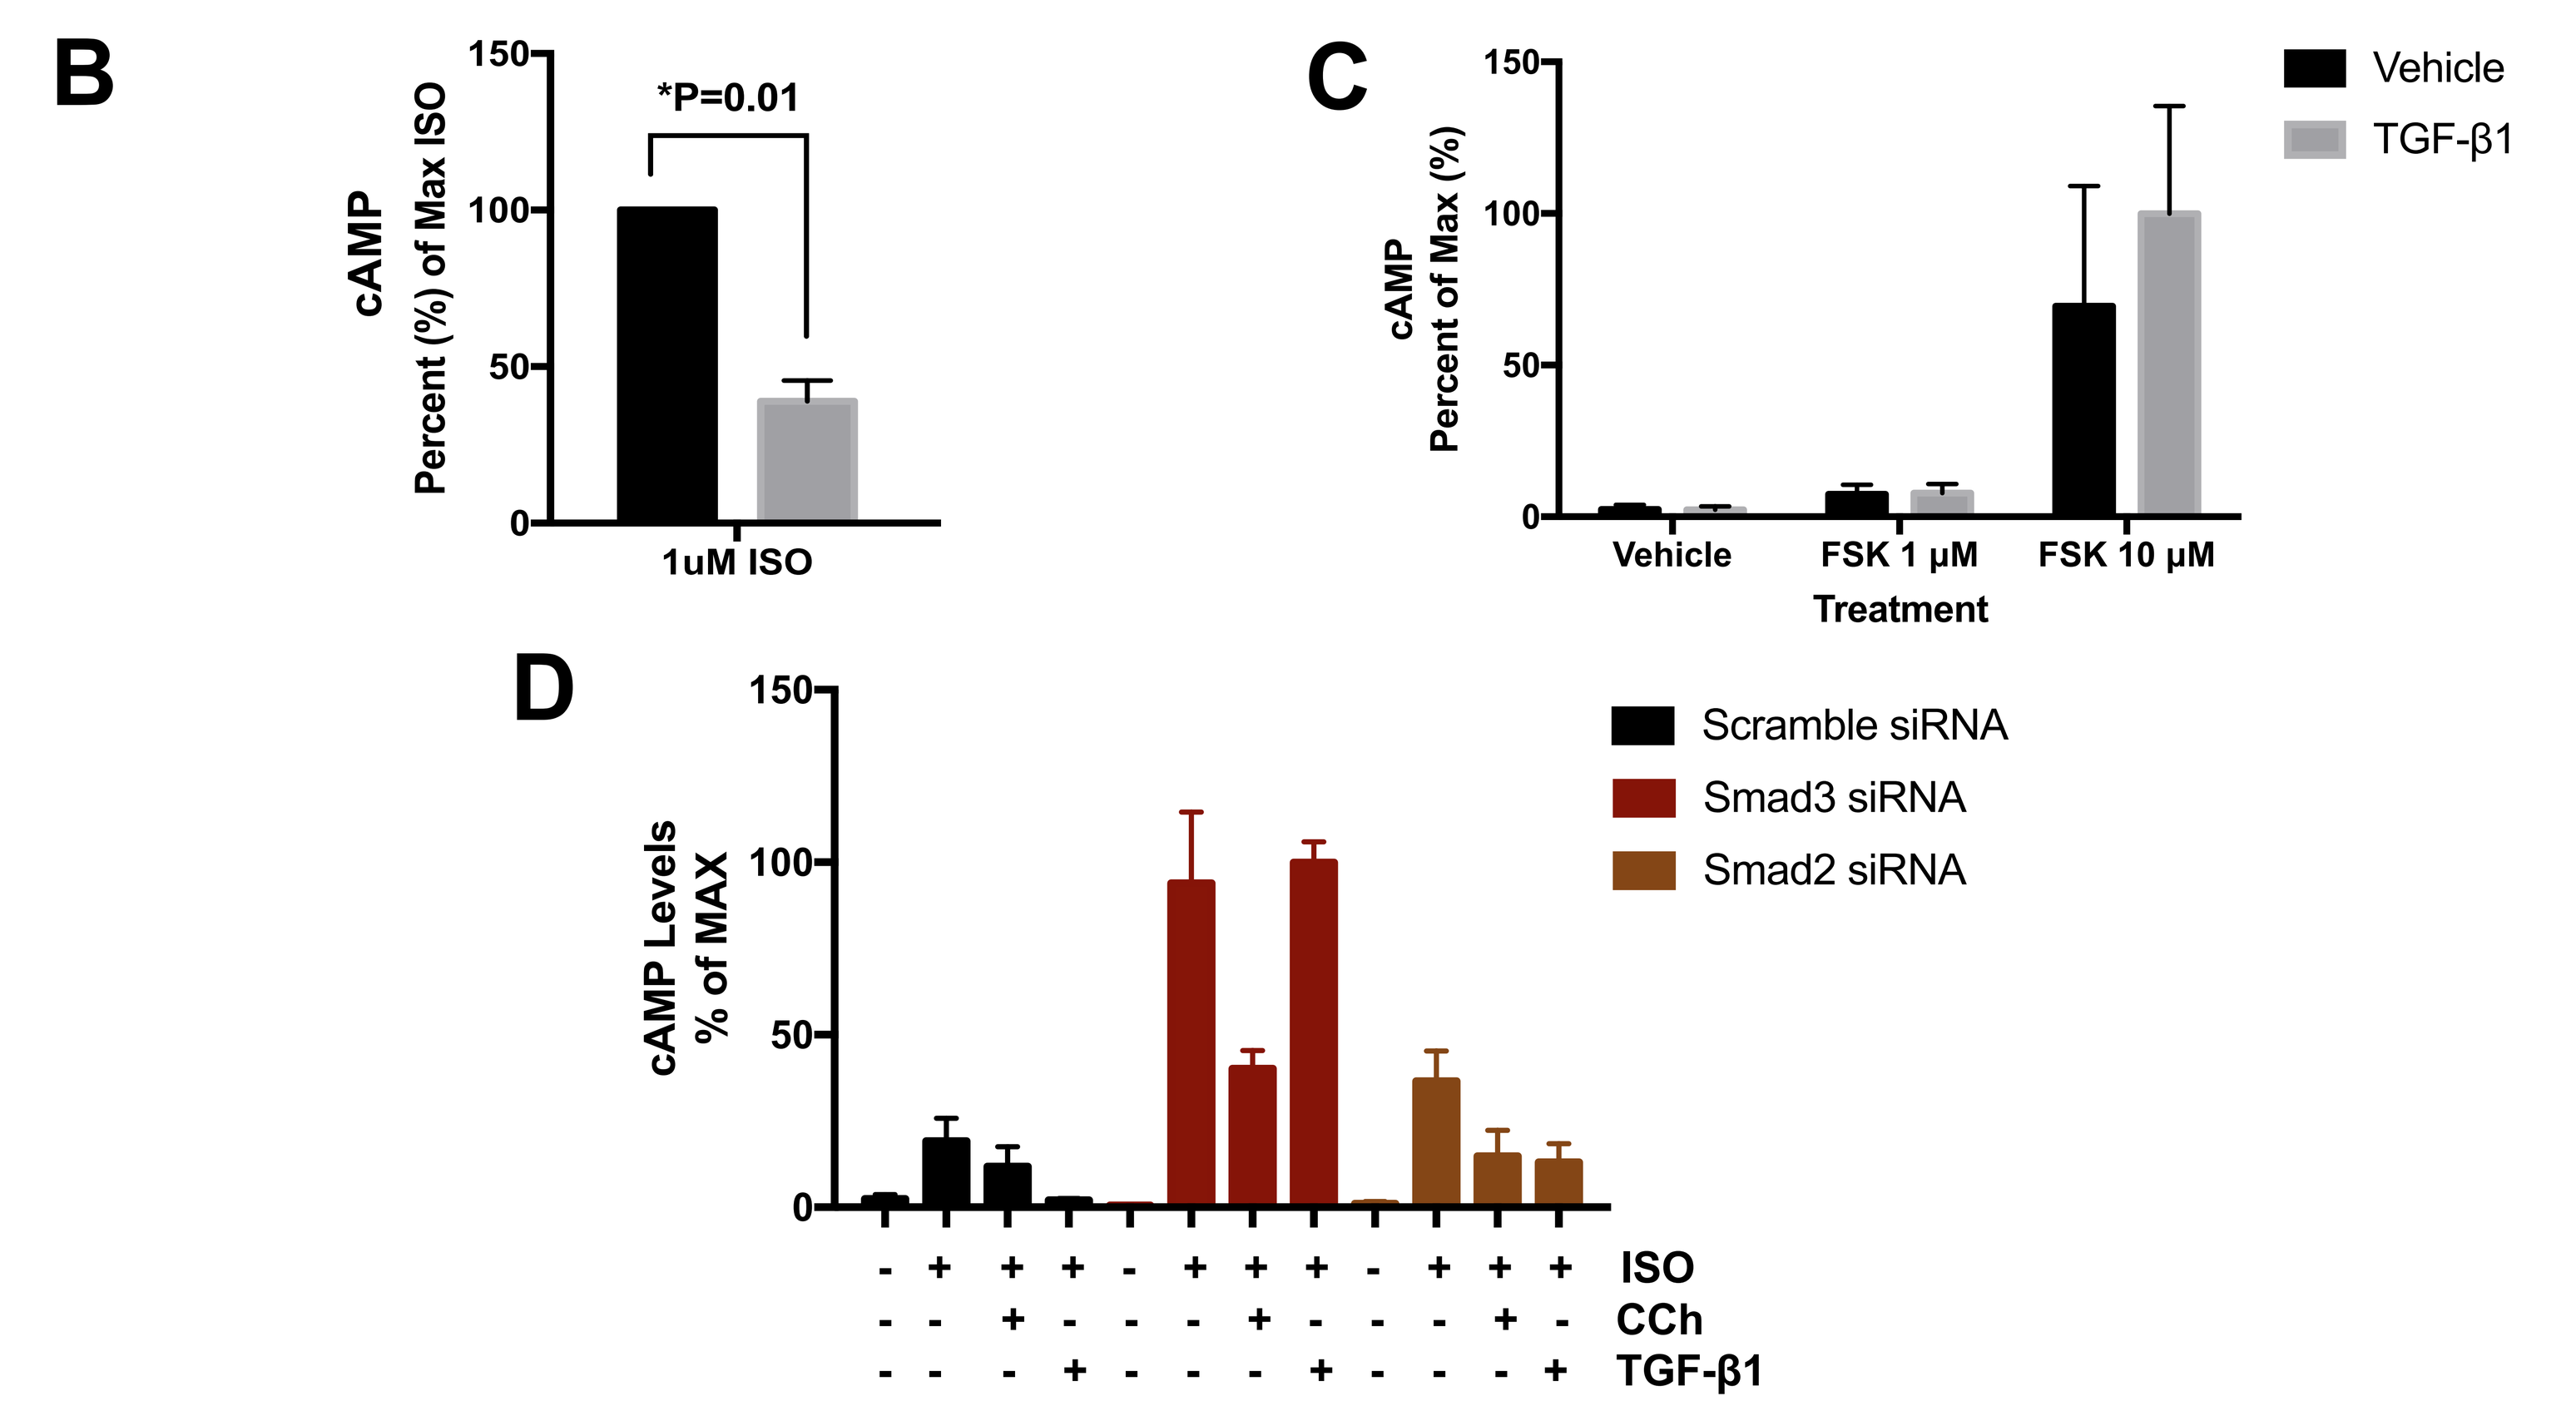

B
cAMP Percent (%) of Max ISO
*P=0.01
1uM ISO
C
cAMP Percent of Max (%)
Vehicle
TGF-β1
FSK 1 μM
FSK 10 μM
Treatment
D
cAMP Levels % of MAX
Scramble siRNA
Smad3 siRNA
Smad2 siRNA
ISO
CCh
TGF-β1

Figure 1. TGF-β1 Inhibits HASM Cell Responsiveness to β2AR Stimuli. A) Phosphorylated MLC after TGF-β1 (10 ng/mL, 18h), carbachol (Cch, 20 μM), and/or isoproterenol (ISO, 1 μM) treatment. **B)** TGF-β1 effect on ISO and **C)** forskolin (FSK)-induced cAMP **D)** in siRNA-transfected HASM cells. **P ≤ 0.05; N=4-8 +/-SEM*

TGF-β1 Induces β2-Agonist Resistance in Isolated HASM Cells

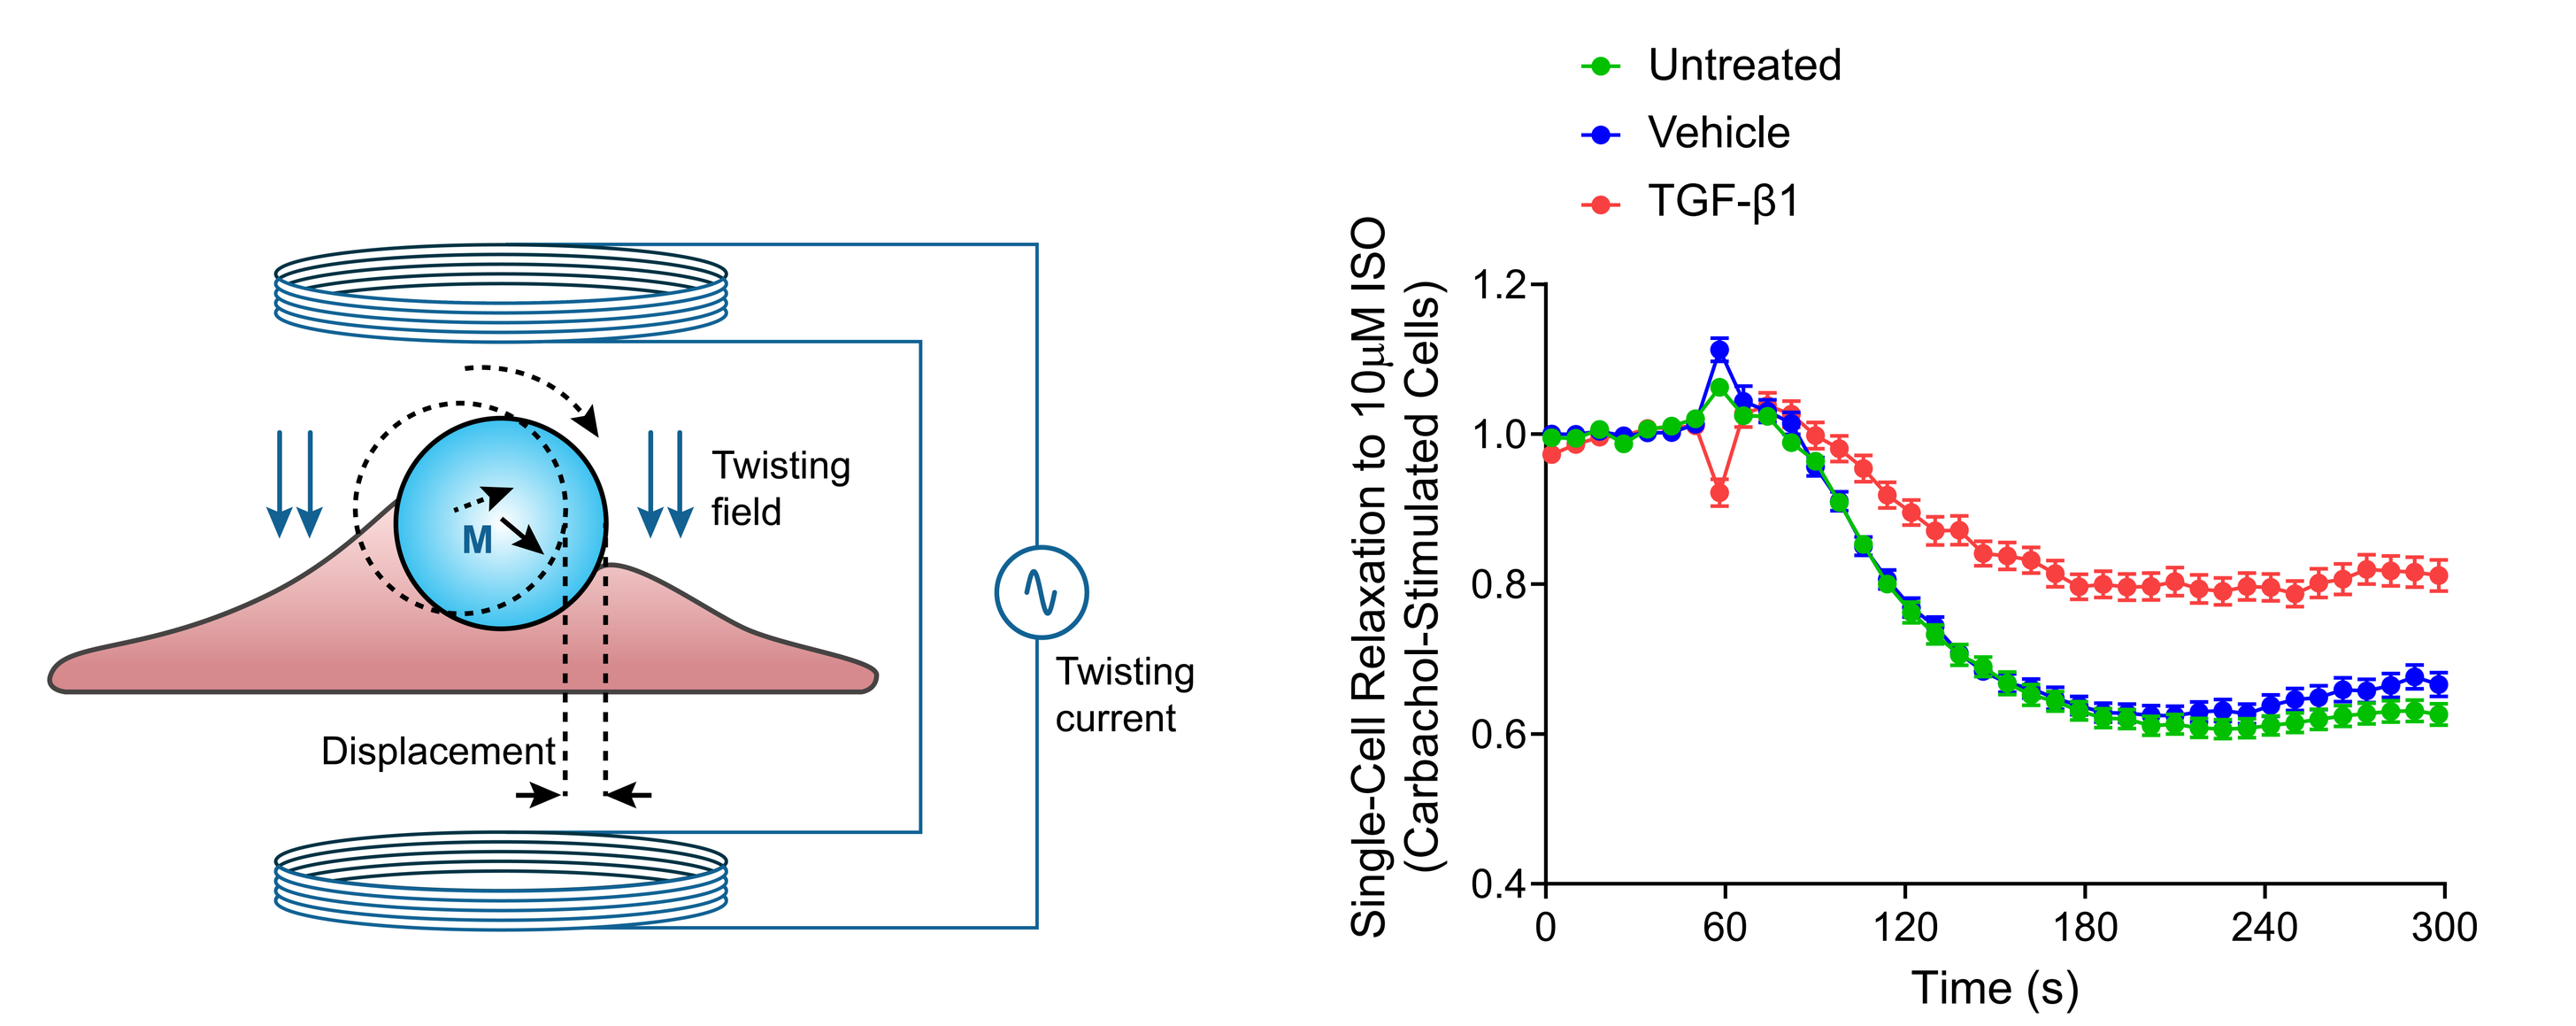

Twisting field
Twisting current
Displacement
M
Untreated
Vehicle
TGF-β1
Single-Cell Relaxation to 10μM ISO (Carbachol-Stimulated Cells)
Time (s)

Figure 2. TGF-β1 Induces β2-Agonist Resistance in Isolated HASM Cells. Isolated HASM cells treated with TGF-β1 (10 ng/mL, 24 h) pre-contracted to Cch, and stimulated with ISO *(N=480-636; +/-SD).*

TGF-β1 Modulates β2-Agonist-Induced hPCLS Relaxation

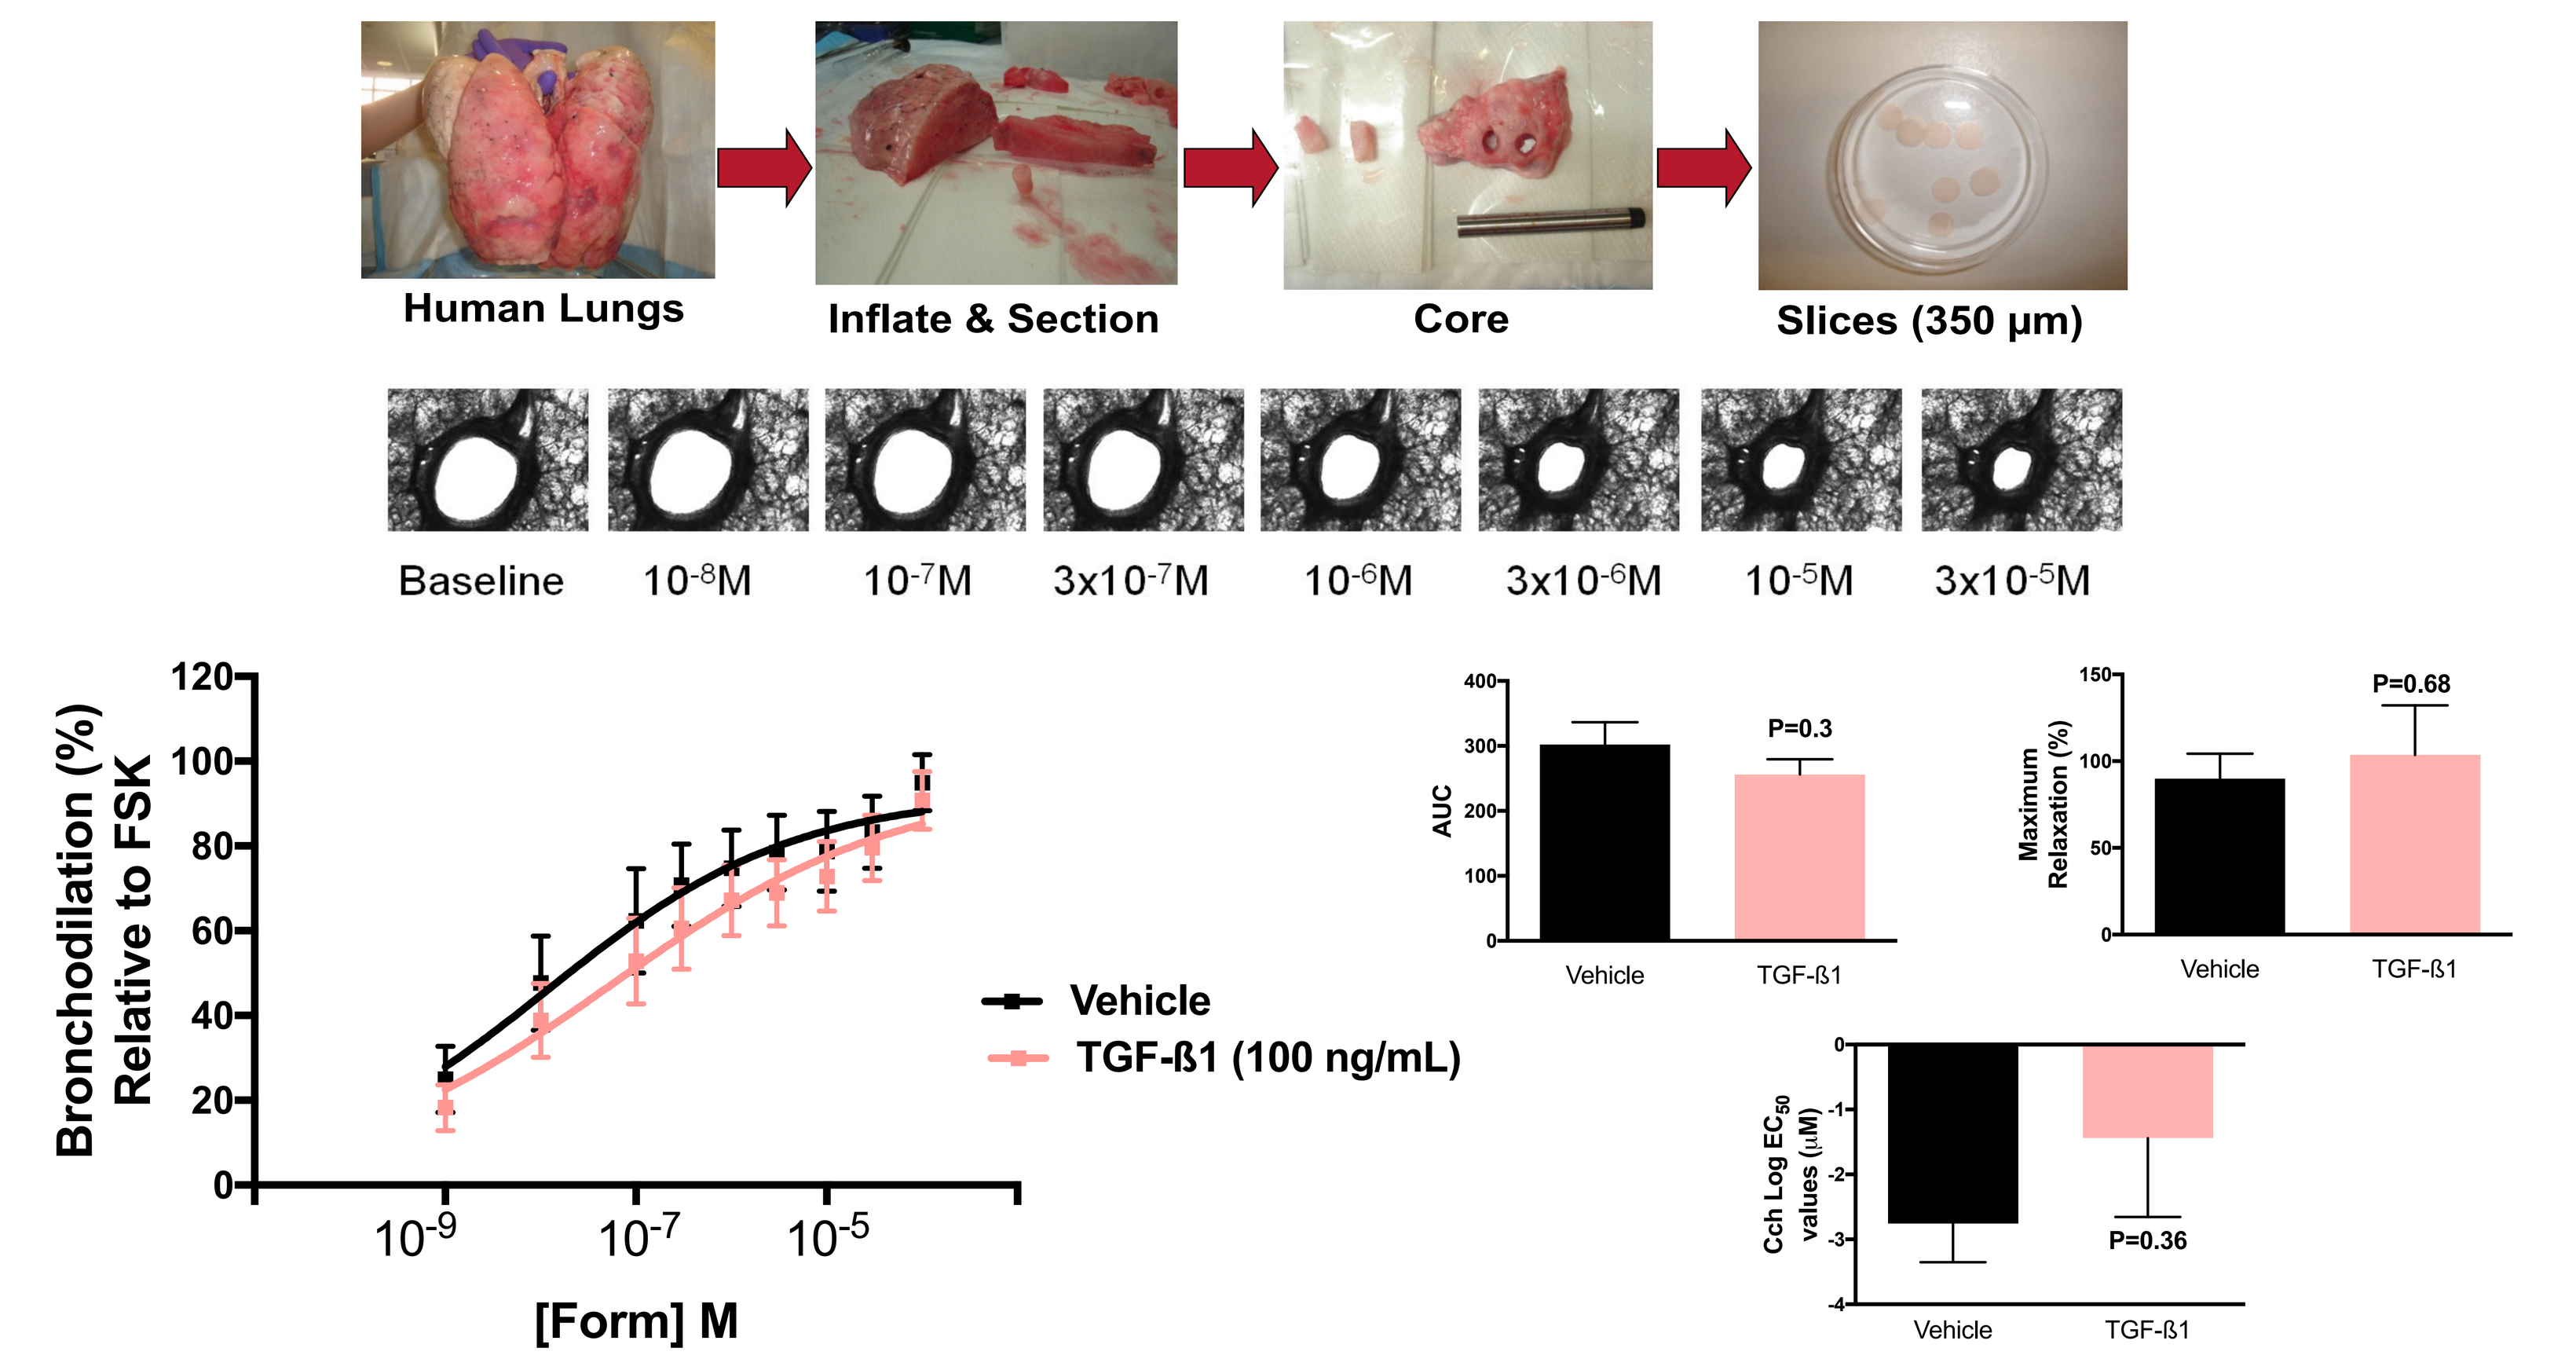

Human Lungs
Inflate & Section
Core
Slices (350 μm)
Baseline
10-8M
10-7M
3x10-7M
10-6M
3x10-6M
10-5M
3x10-5M
Bronchodilation (%) Relative to FSK
[Form] M
Vehicle
TGF-ß1 (100 ng/mL)
AUC
P=0.3
Maximum Relaxation (%)
P=0.68
Cch Log EC50 values (μM)
P=0.36

Figure 3. TGF-β1 Modulates β2-Agonist-Induced hPCLS Relaxation. hPCLS were treated with TGF-β1 (18 h, 100 ng/mL), pre-contracted to Cch, and subjected to a Formoterol (Form) dose response (N=7; +/- SEM)

Phosphodiesterase Inhibition Reverses TGF-β1 Effects on β2AR Function

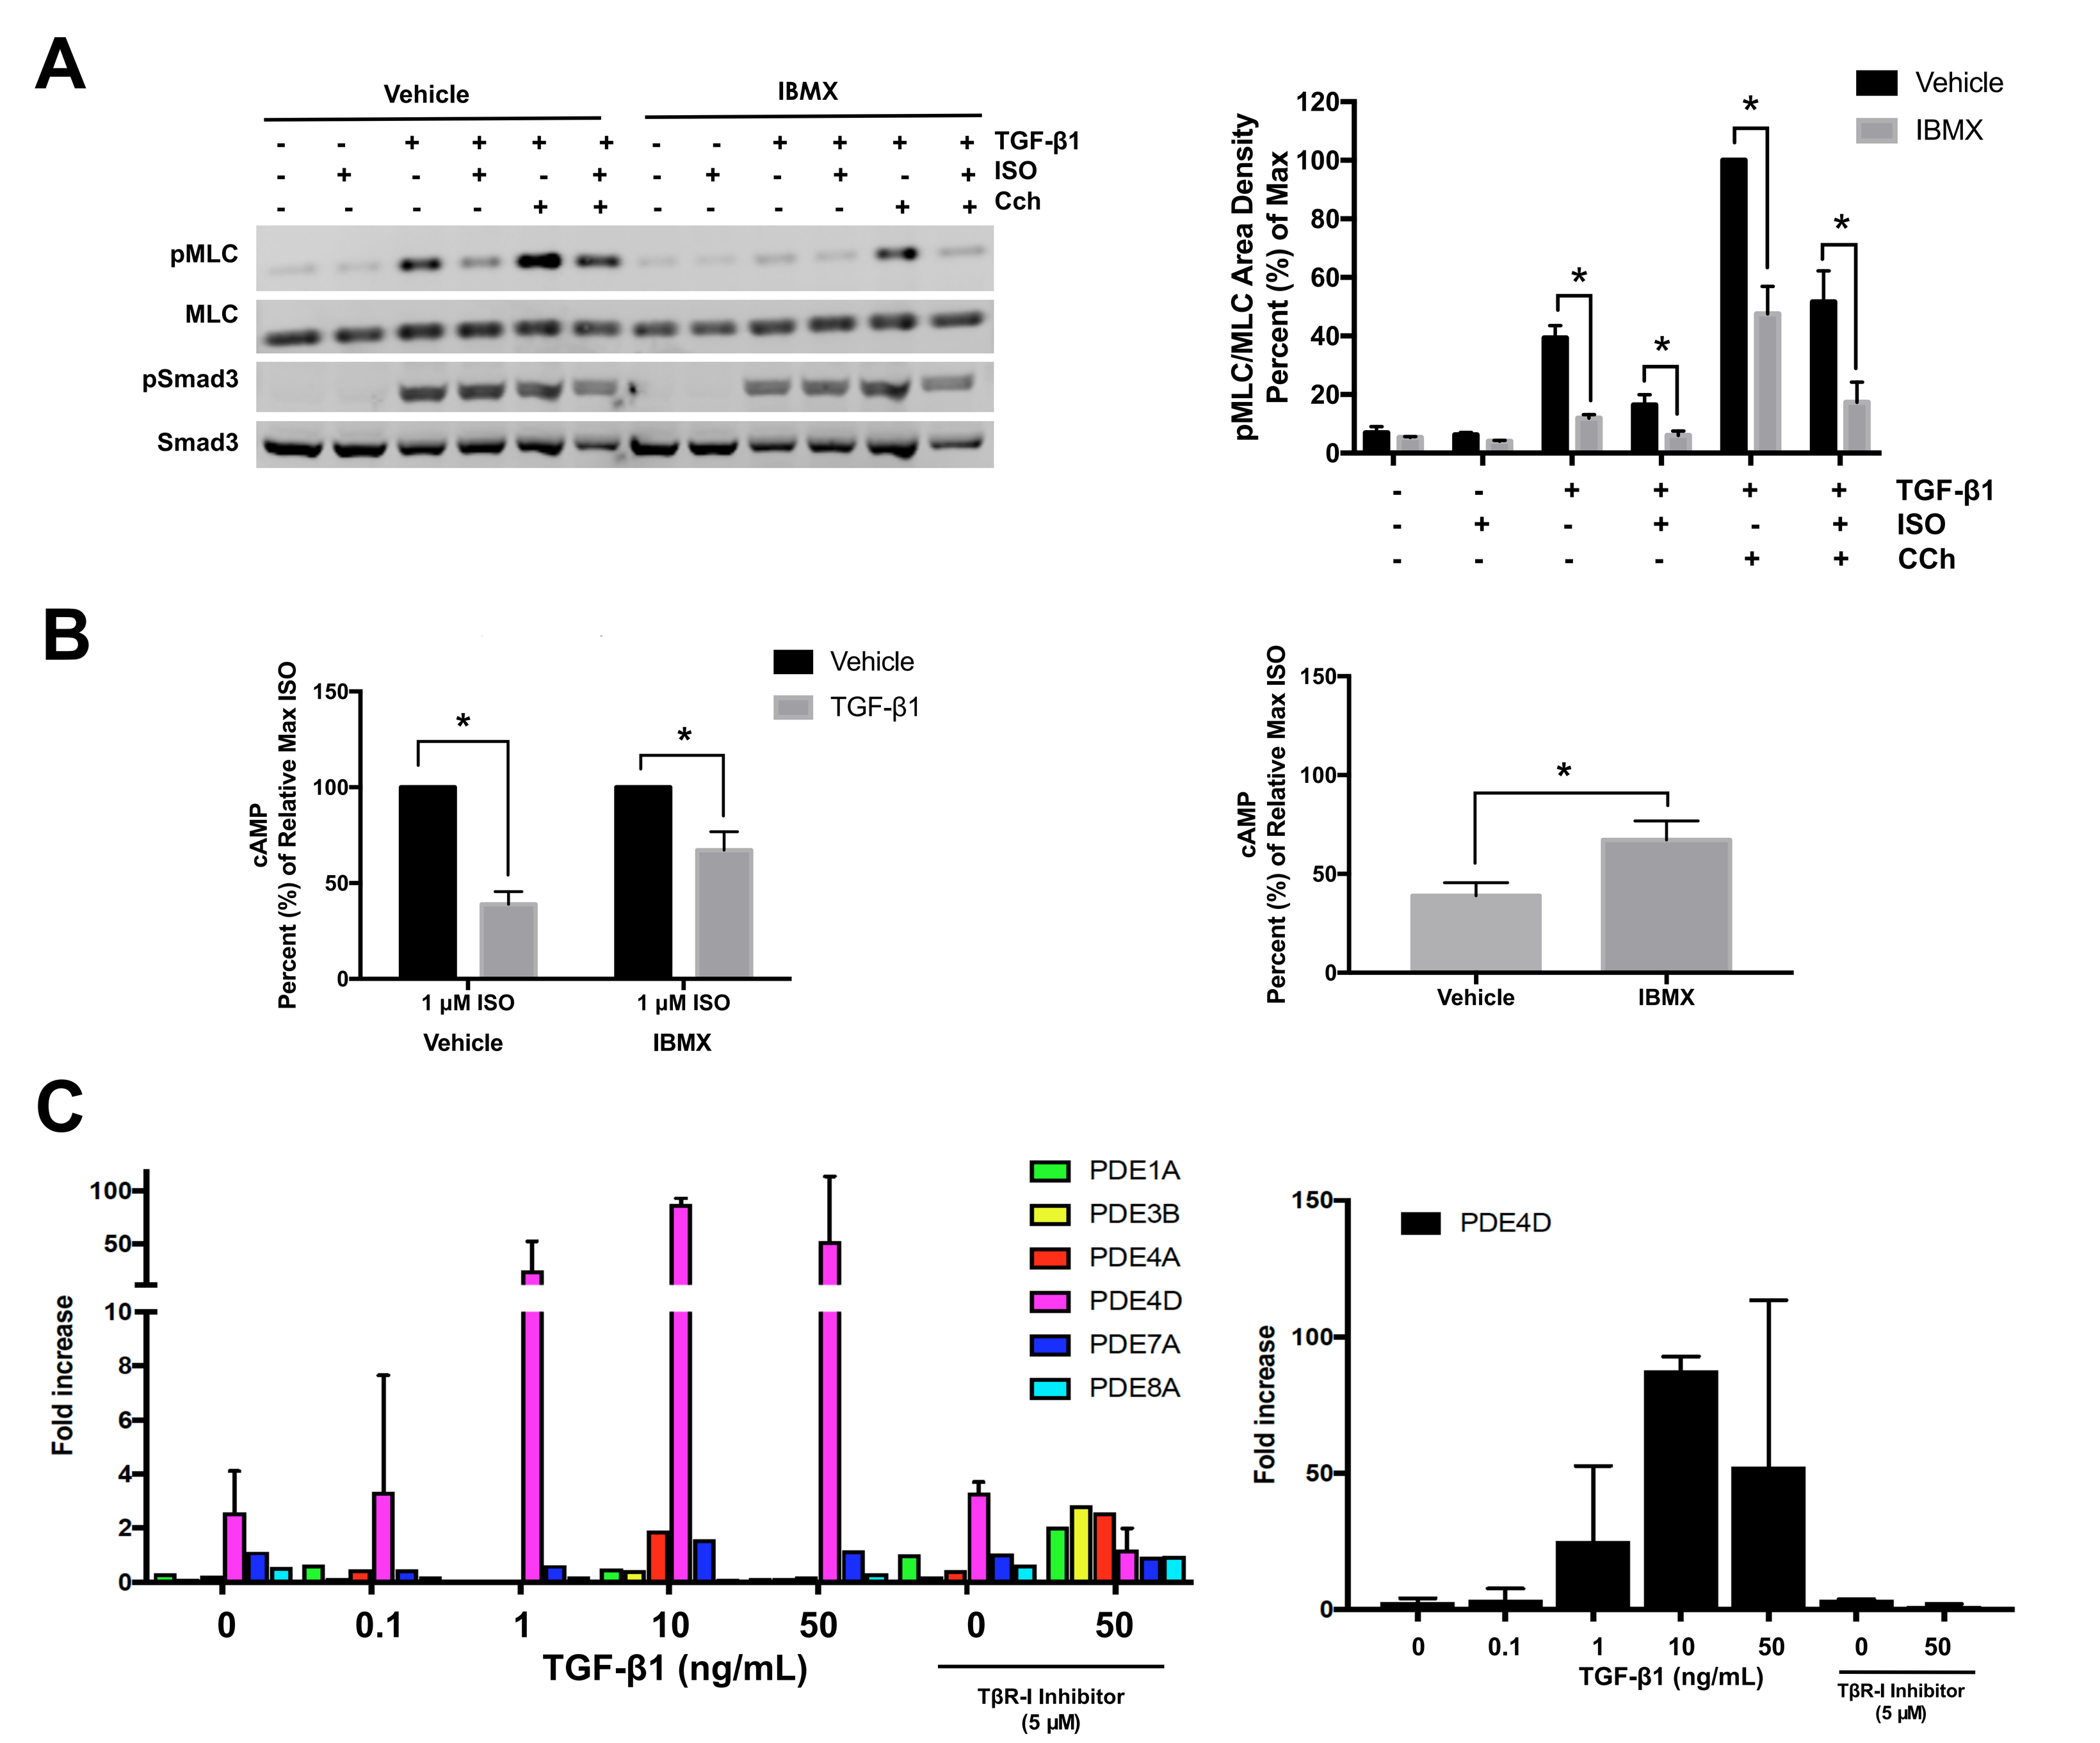

A
Vehicle
IBMX
TGF-β1
ISO
Cch
pMLC
MLC
pSmad3
Smad3
pMLC/MLC Area Density Percent (%) of Max
CCh
B
cAMP Percent (%) of Relative Max ISO
TGF-β1
1 μM ISO
C
Fold increase
PDE1A
PDE3B
PDE4A
PDE4D
PDE7A
PDE8A
TGF-β1 (ng/mL)
TβR-I Inhibitor (5 μM)

Figure 4. Phosphodiesterase Inhibition Reverses TGF-β1 Effects on β2AR Function. A) MLC phosphorylation (N=3 +/-SEM) and **B)** cAMP generation (N=6-8+/-SEM) in Cch, ISO, TGF-β1 (24 h, 10 ng/mL), and IBMX pre-treated HASM cells. **C)** PDE gene expression in TGF-β1-treated HASM cells. **P ≤ 0.05*

SUMMARY

- TGF-β1 diminishes ISO-induced inhibition of MLC phosphorylation
- TGF-β1 diminishes ISO-induced cAMP generation
- Smad3 inhibition reverses TGF-β1's effects on ISO-induced cAMP
- TGF-β1 induces β2-agonist resistance in isolated HASM cells
- TGF-β1 modulates β2-agonist-induced hPCLS relaxation
- TGF-β1 induces PDE4D gene expression
- Phosphodiesterase inhibition reverses TGF-β1 effects on β2AR function

SIGNIFICANCE

- Novel role for TGF-β1 in modulating HASM cell β2AR function
- We speculate that TGF-β1 release following repeated airway injury-repair may increase bronchomotor tone and β2-agonist insensitivity

REFERENCES

1. Ojiaku CA, Yoo EJ, Panettieri RA Jr.. Transforming Growth Factor β1 Function in Airway Remodeling and Hyperresponsiveness. The Missing Link? Am J Respir Cell Mol Biol. 2017 Apr;56(4):432-442.
2. Ojiaku CA, Cao G, Zhu W, Jude JA, Yoo EJ, An SS, Panettieri RA, Jr. TGF-β1 Human Airway Smooth Muscle Shortening is Smad3-Dependent. Am J Respir Cell Mol Biol. 2017 Oct 6. [Epub ahead of print]
3. Cooper, P. R. & Panettieri, R. A. Steroids completely reverse albuterol-induced beta(2)-adrenergic receptor tolerance in human small airways. J. Allergy Clin. Immunol. 122, 734–40 (2008).

ACKNOWLEDGEMENTS

These studies were supported by a NIH Predoctoral Training Grant in Pharmacology (5-T32-GM-008076-28)
&
NIH/NHLBI 1P01-HL114471-01A1

QR Code
Rutgers Institute for Translational Medicine and Science